Identifying FGFR2 *fusions/rearrangements in cholangiocarcinoma patients using a novel cfDNA algorithm for treatment with RLY-4008, a highly selective irreversible FGFR2 inhibitor*

Alison M. Schram[1], Mitesh Borad[2], Vaibhav Sahai[3], Suneel Kamath[4], Richard Kim[5], Chih-Yi Andy Liao[6], Do-Youn Oh[7], Mariano Ponz-Sarvisé[8], Jeffrey Yachnin[9], Scott A. Shell[10], Philippe Cassier[11], Efrat Dotan[12], Vaia Florou[13], Victor Moreno[14], Joon Oh Park[15], David Tai[16], Oleg Schmidt-Kittler[17], Charles Ferté[17], Lipika Goyal[18], Vivek Subbiah[19]

[1]Memorial Sloan Kettering Cancer Center, New York, USA; [2]Mayo Clinic Scottsdale – PPDS, Phoenix, Arizona, USA; [3]University of Michigan, Ann Arbor, Michigan, USA; [4]The Cleveland Clinic Taussig Cancer Institute, Cleveland, Ohio, USA; [5]H Lee Moffitt Cancer Center and Research Institute, Tampa, Florida, USA; [6]University of Chicago Medical Center, Chicago, Illinois, USA; [7]Seoul National University Hospital, Seoul, Republic of Korea; [8]Clinica Universidad Navarra, Pamplona, Spain; [9]Karolinska Universitetssjukhuset Solna, Stockholm, Sweden; [10]Guardant Health, Palo Alto, California, USA; [11]Centre Léon Bérard Centre, Lyon, France; [12]Fox Chase Cancer Center, Philadelphia, Pennsylvania, USA; [13]University of Utah – Huntsman Cancer Institute – PPDS, Salt Lake City, Utah, USA; [14]START MADRID Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [15]Samsung Medical Center, Sungkyunkwan University School of Medicine, Seoul, Republic of Korea; [16]National Cancer Centre, Singapore; [17]Relay Therapeutics, Cambridge, Massachusetts, USA; [18]Mass General Cancer Center, Boston, Massachusetts, USA; [19]University of Texas MD Anderson Cancer Center, Houston, Texas, USA

RELAY® THERAPEUTICS

Poster 326

BACKGROUND

- Cholangiocarcinoma (CCA) is an aggressive malignancy with a dismal prognosis, typically treated with chemotherapy[1]
- In a subset of patients (~10–15% of intrahepatic CCA cases), CCA tumours harbour *FGFR2* fusions/rearrangements (f/r) that drive tumour growth[1]
- Identification of these patients is crucial since they are likely to benefit from FGFR2-targeted therapy
- However, genomic profiling based on tumour biopsy can pose challenges, with limited tumour tissue available, and a tendency to forego repeat biopsies in favour of initiating therapy due to the aggressive nature of CCA
- Liquid biopsies may be a non-invasive way to identify patients most likely to benefit from FGFR2-targeted therapy. Recent technical advances in cell-free DNA (cfDNA) analysis have increased the sensitivity of this method for detecting *FGFR2* f/r[2]
- We evaluated the sensitivity of liquid biopsies in detecting *FGFR2* f/r, compared to local and central tissue assessments, in patients from the ReFocus study (NCT04526106)[3]
 - ReFocus is a study of RLY-4008, a potent, selective, specific, and irreversible FGFR2 inhibitor (FGFRi),[4] in patients with advanced, *FGFR2*-driven CCA or other solid tumours. It has shown promising efficacy to date in FGFRi-naïve patients with *FGFR2* f/r CCA **(Figure 1)**[5]

Figure 1. Radiographic tumour regression per RECIST 1.1 – RP2D[5]

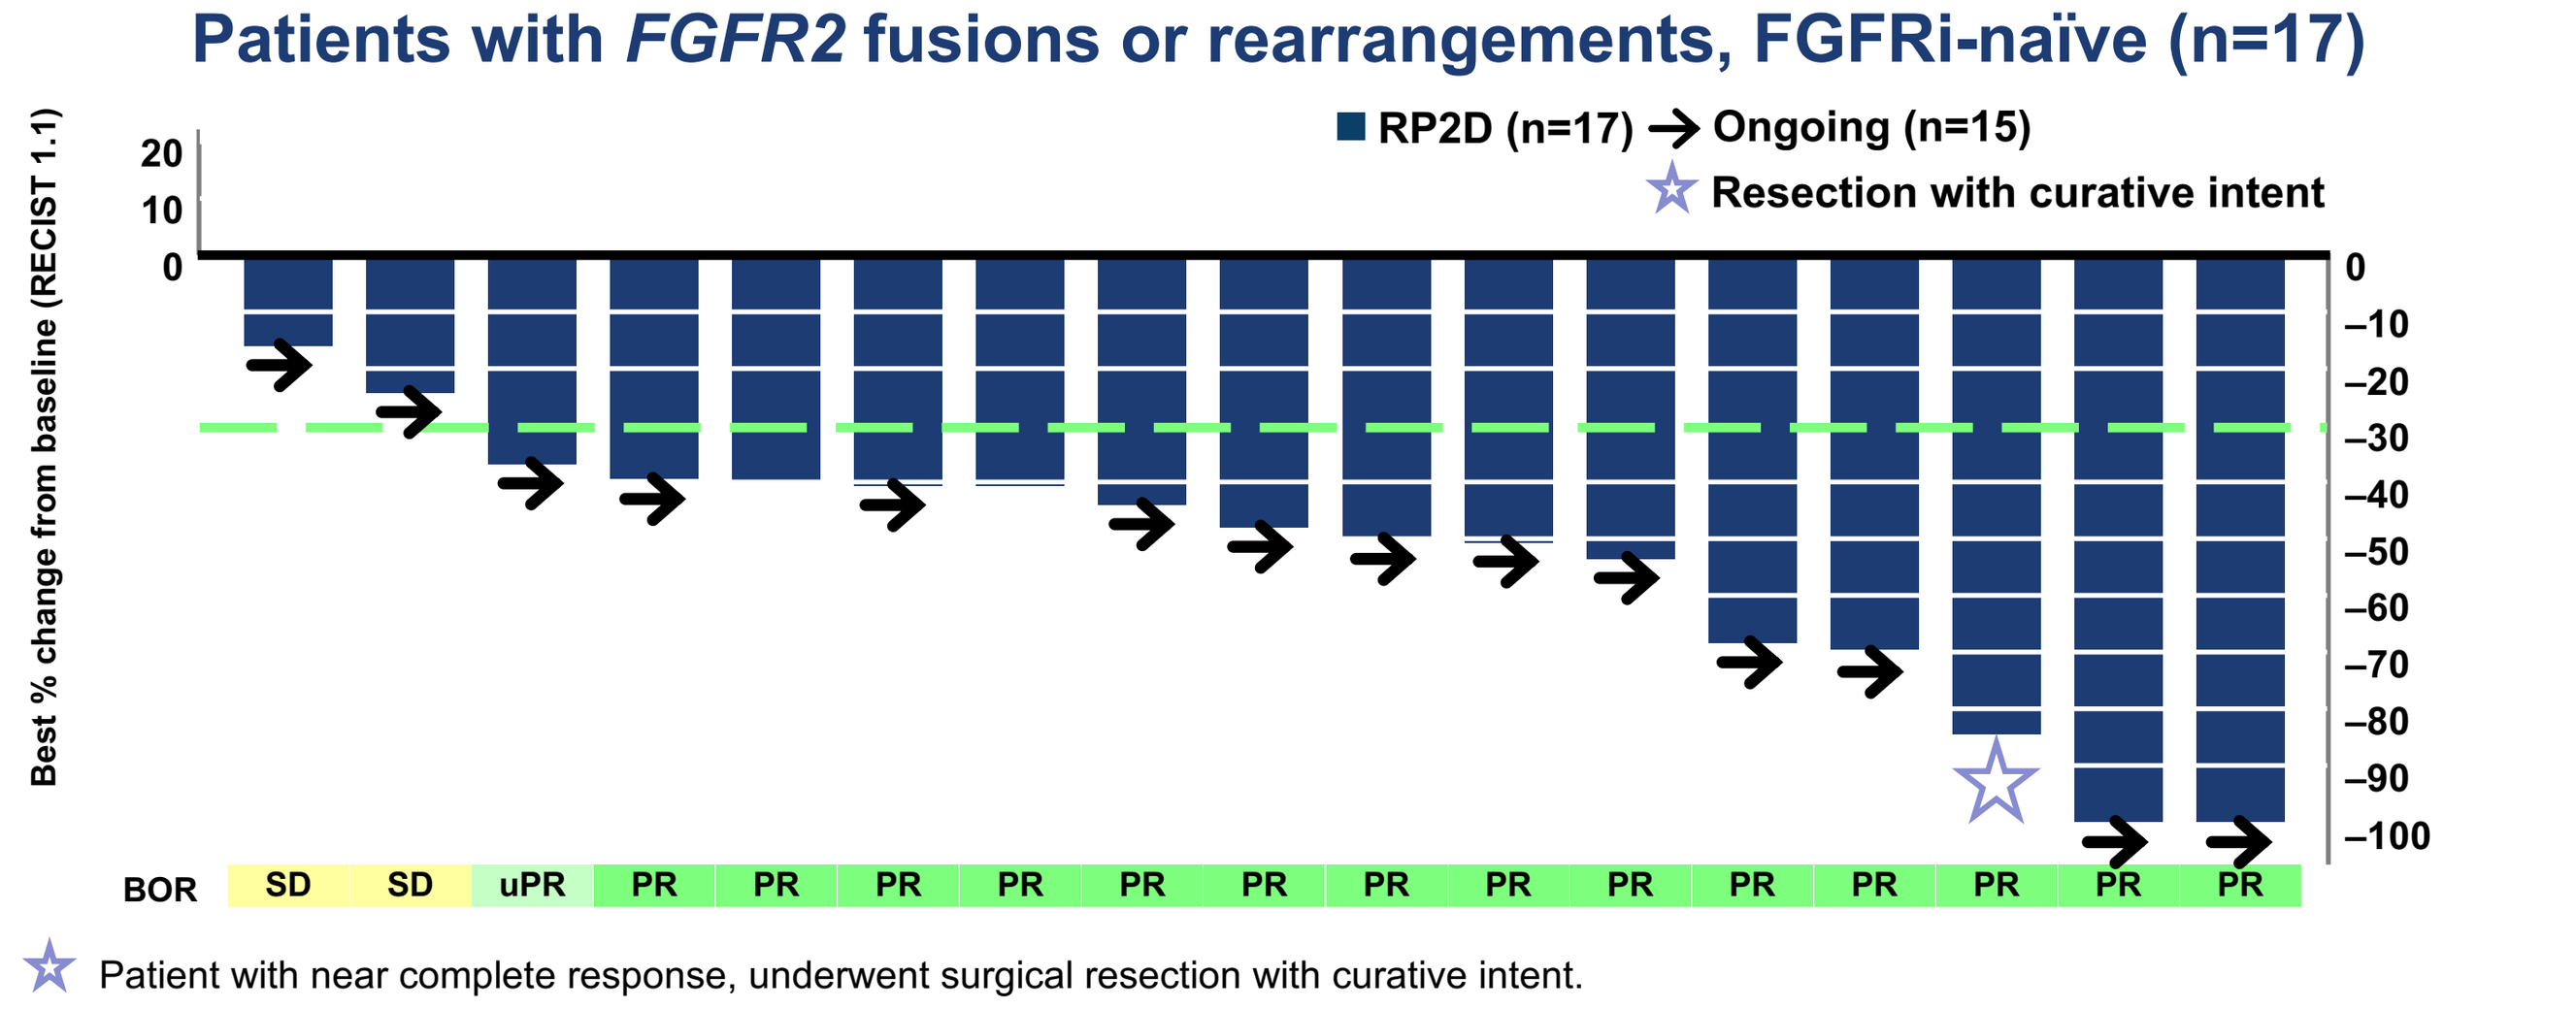
Patients with *FGFR2* fusions or rearrangements, FGFRi-naïve (n=17)
RP2D (n=17) → Ongoing (n=15)
Resection with curative intent
Best % change from baseline (RECIST 1.1)
20
10
0
0
–10
–20
–30
–40
–50
–60
–70
–80
–90
–100
BOR
SD SD uPR PR PR PR PR PR PR PR PR PR PR PR PR PR PR

☆ Patient with near complete response, underwent surgical resection with curative intent.

BOR, best objective response; FGFRi, FGFR inhibitor; (u)PR, (unconfirmed) partial response; RP2D, recommended Phase 2 dose; SD, stable disease, RECIST, Response Evaluation Criteria In Solid Tumours.

METHODS

Samples

- Tumour tissue (archival or fresh biopsies) and pre-treatment plasma samples were obtained for central analysis from 73 patients in the ReFocus trial, until 8 September, 2022 (data cut-off)

Objective

- To evaluate the feasibility (assessed according to failure rate) and the sensitivity (assessed according to percent positive agreement) of liquid biopsies to identify *FGFR2* f/r, compared to local and central tissue assessments

Sample analysis for *FGFR2* f/r

- Pre-treatment samples were analysed centrally, and methods applied per **Figure 2**
- Liquid cfDNA samples were then analysed *in silico* using a fusion partner agnostic algorithm that is able to detect unique *FGFR2* f/r
- Details of the Guardant360® (G360) research-only algorithm have been described previously.[2] This algorithm was applied to the G360 test to broaden the capability of detecting *FGFR2* f/r

Patient demographics

- Of the 73 patients who provided samples, 40% were male; the mean age was 56 years (standard deviation: 14 years)
- The majority of patients (93%) had Eastern Cooperative Oncology Group performance status (ECOG PS) of 0 or 1
- Almost all CCA patients (72/73) had intrahepatic disease at diagnosis
- The proportion of patients who had undergone ≥2 prior treatment regimens was 82%
- The median sum of target lesions was 79 mm (range: 13–261 mm)

Figure 2. Diagnostic analysis plan

Total population
- Patients with unresectable/metastatic CCA
- *FGFR2* f/r-positive per local tissue or plasma testing
- Availability of tissue and cfDNA samples

N=73

Samples centrally confirmed f/r positive
Samples *FGFR2* f/r-positive per central tissue (CTA based on FoundationOne® CDx) or central liquid (Guardant360® CDx*)
N=68

Samples positive per central liquid
Samples *FGFR2* f/r-positive per central liquid
N=56

Performance of tissue vs. liquid biopsies (Table 1)
Recall rate, rate of fusion not detected, QNS / no ctDNA rate, PPA

***FGFR2* fusion detection by assay type (Figure 3)**
Venn diagram of *FGFR2* f/r positive, negative, and failed samples

***FGFR2* alterations (Figure 4)**
OncoPrint plot of all identified *FGFR2* alterations by assay

Breakpoint map and partner chromosome (Figure 5)
Circos plot outlining breakpoints and partner genes

***FGFR2* fusion partners (Figure 6)**
Pie chart depicting recurring and unique *FGFR2* f/r partners

*FDA-approved 74-gene cfDNA NGS-based assay
CCA, cholangiocarcinoma; CDx, companion diagnostic; cfDNA, cell-free DNA; ctDNA, circulating tumour DNA; CTA, clinical trial assay; *FGFR2* f/r, *FGFR2* fusions/rearrangements; QNS, quantity not sufficient; NGS, next-generation sequencing; PPA, percent positive agreement.

Figure 3. *FGFR2* f/r detection by assay type

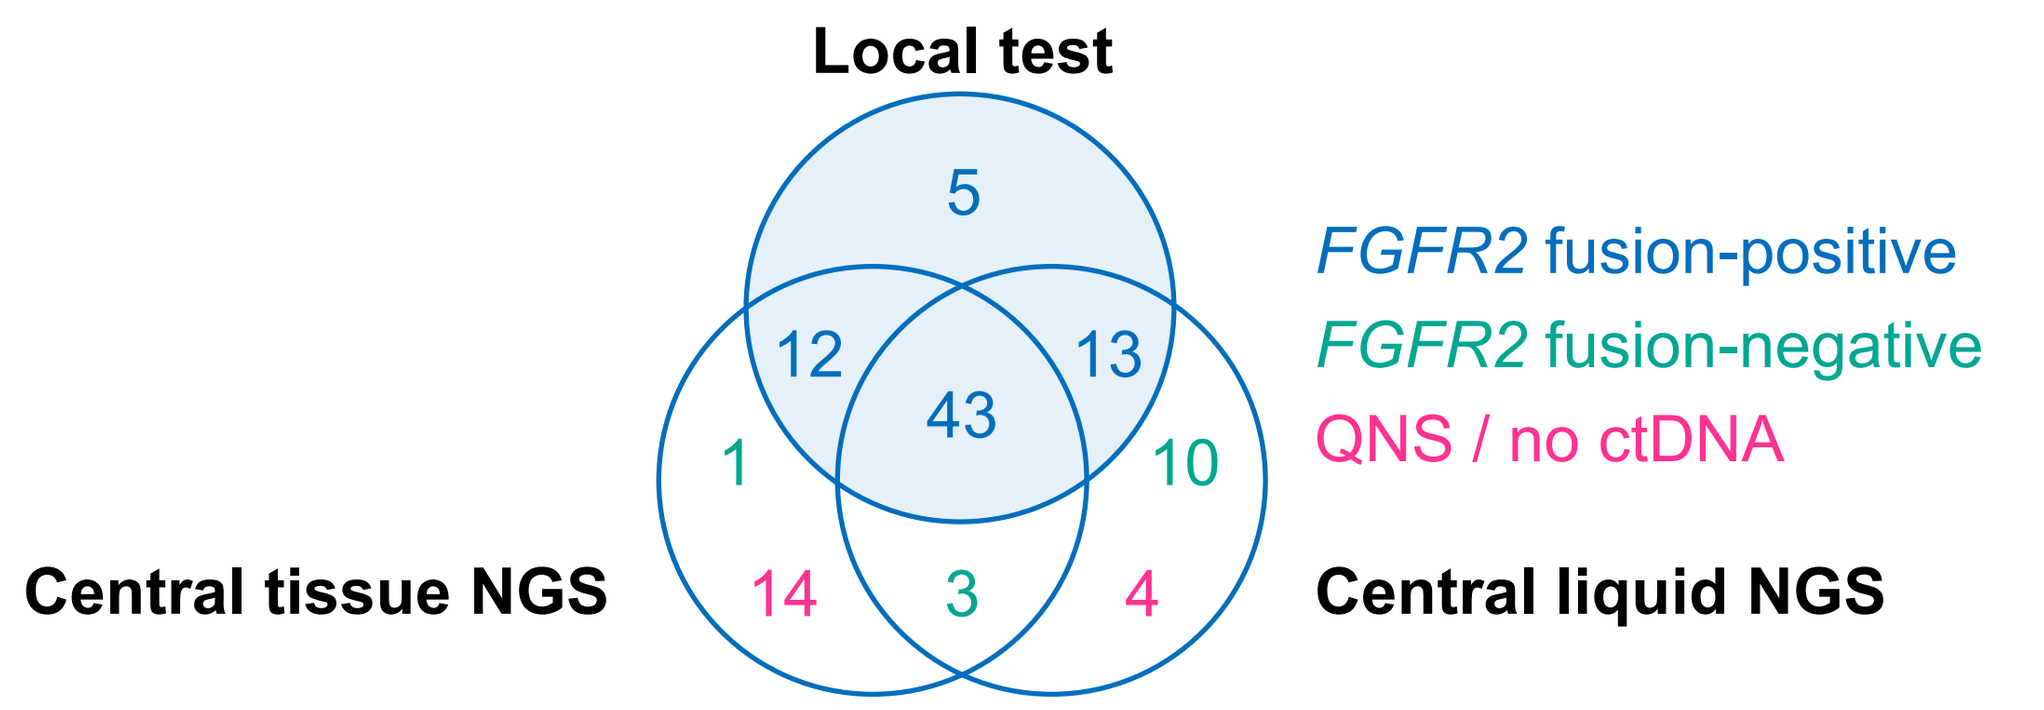
Local test
5
12
13
43
1
10
14
3
4
Central tissue NGS
Central liquid NGS
FGFR2 fusion-positive
FGFR2 fusion-negative
QNS / no ctDNA

ctDNA, circulating tumour DNA; *FGFR2* f/r, *FGFR2* fusions/rearrangements; QNS, quantity not sufficient; NGS, next-generation sequencing.

RESULTS

Percentage of *FGFR2* fusions detected in tissue vs. liquid biopsies

- Of the 73 tumour tissue samples, central tissue next-generation sequencing (NGS) detected 55 *FGFR2* f/r (75.3%)
- Of the 73 plasma samples, central liquid NGS detected 56 *FGFR2* f/r (76.7%):
 - 32 *FGFR2* f/r (43.8%) were detected using the standard algorithm
 - The partner agnostic algorithm detected 24 additional *FGFR2* f/r (32.8%)
 - The majority of *FGFR2* f/r identified using the partner agnostic algorithm were unique (18/24, 75%), in contrast to 9/32 (28.1%) using the standard algorithm

Figure 4. *FGFR2* alterations

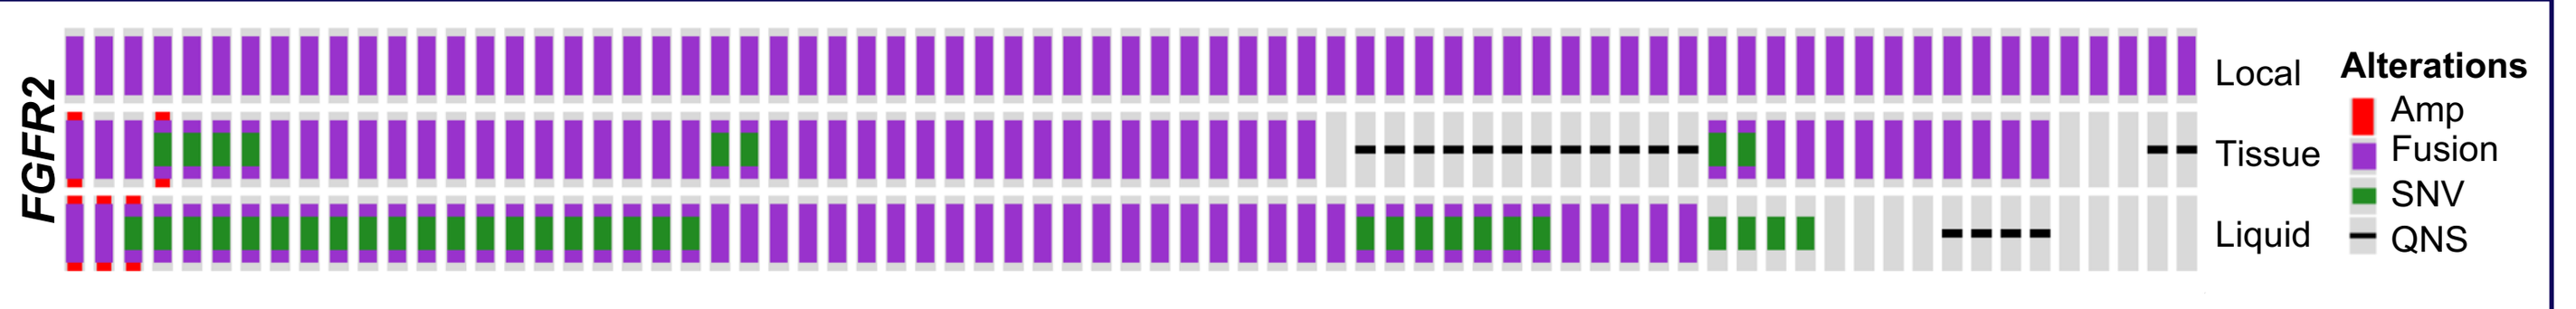
FGFR2
Local
Tissue
Liquid
Alterations
Amp
Fusion
SNV
QNS

Amp, amplification; QNS, quantity not sufficient; SNV, single nucleotide variant. Credit: ComplexHeatmap r package[6]

FGFR2 alterations

- As shown in **Figure 4**, small nucleotide variants (SNV) were the second most common *FGFR2* alterations after *FGFR2* f/r
 - Liquid biopsies identified 31 patients with *FGFR2* SNVs, whereas tissue biopsy identified eight
- Analysis of liquid biopsies detected three amplifications; two amplifications were detected in tissue biopsies. All detected amplifications were low level with a copy number <4
 - Copy number determined by liquid biopsy cannot be directly compared to tissue
- Concordance of *FGFR2* SNVs and amplifications in tissue and liquid was 35/55 (63.6%), much lower than the concordance for *FGFR2* fusions, which was 46/55 (83.6%)
 - 31/33 patients with *FGFR2* SNVs had prior treatment with an FGFRi, suggesting emergence of acquired resistance as a potential cause of the observed discordance

Table 1. Performance of tissue vs. liquid biopsies

	Local tests compared to:			Central tissue compared to:	
	Central tissue	Central liquid (standard)	Central liquid (partner agnostic)	Central liquid (standard)	Central liquid (partner agnostic)
Recall rate*	55/73 (75.3%)	32/73 (43.8%)	56/73 (76.7%)	25/55 (45.5%)	43/55 (78.2%)
Fusion not detected rate†	4/73 (5.5%)	37/73 (50.7%)	13/73 (17.8%)	26/55 (47.3%)	8/55 (14.5%)
QNS / no ctDNA rate‡	14/73 (19.2%)	4/73 (5.5%)		4/55 (7.3%)	
Percent positive agreement**	55/59 (93.2%)	32/69 (46.4%)	56/69 (81.2%)	25/55 (45.5%)	46/55 (83.6%)

*Recall rate = *FGFR2* f/r-positive cases by assay / total population; †Fusion not detected = successfully reported samples without *FGFR2* f/r call / total population; ‡QNS / no ctDNA rate = samples failed QC or no ctDNA / total population; **Percent positive agreement = matched results / number of successfully reported samples.

ctDNA, circulating tumour DNA; *FGFR2* f/r, *FGFR2* fusions/rearrangements; QC, quality control; QNS, tumour quantity not sufficient.

FGFR2 fusion detection by biopsy and assay type

- In 68 patients, tissue or liquid biopsies identified *FGFR2* fusion events **(Figure 3; Table 1)**
 - In 43 patients, the local and central NGS tests detected identical FGFR2 f/r events
- Five *FGFR2* fusions identified in local tests were not confirmed by the central tests
 - Three patients were *FGFR2* fusion-negative according to results of both tumour tissue and liquid biopsy analyses
 - Two patients were negative by central liquid biopsy and tumour 'quantity not sufficient' (QNS) in central tissue NGS
- The frequency of central NGS *FGFR2* f/r negativity was similar for both biopsy types but differed in reason:
 - Analysis of tissue biopsy resulted in a higher rate of pathology QNS (14/73)
 - Analysis of liquid biopsy resulted in a higher rate of *FGFR2* fusion-negative outputs (10/73)

Figure 5. Breakpoint map and partner chromosomes (tissue and liquid biopsies)

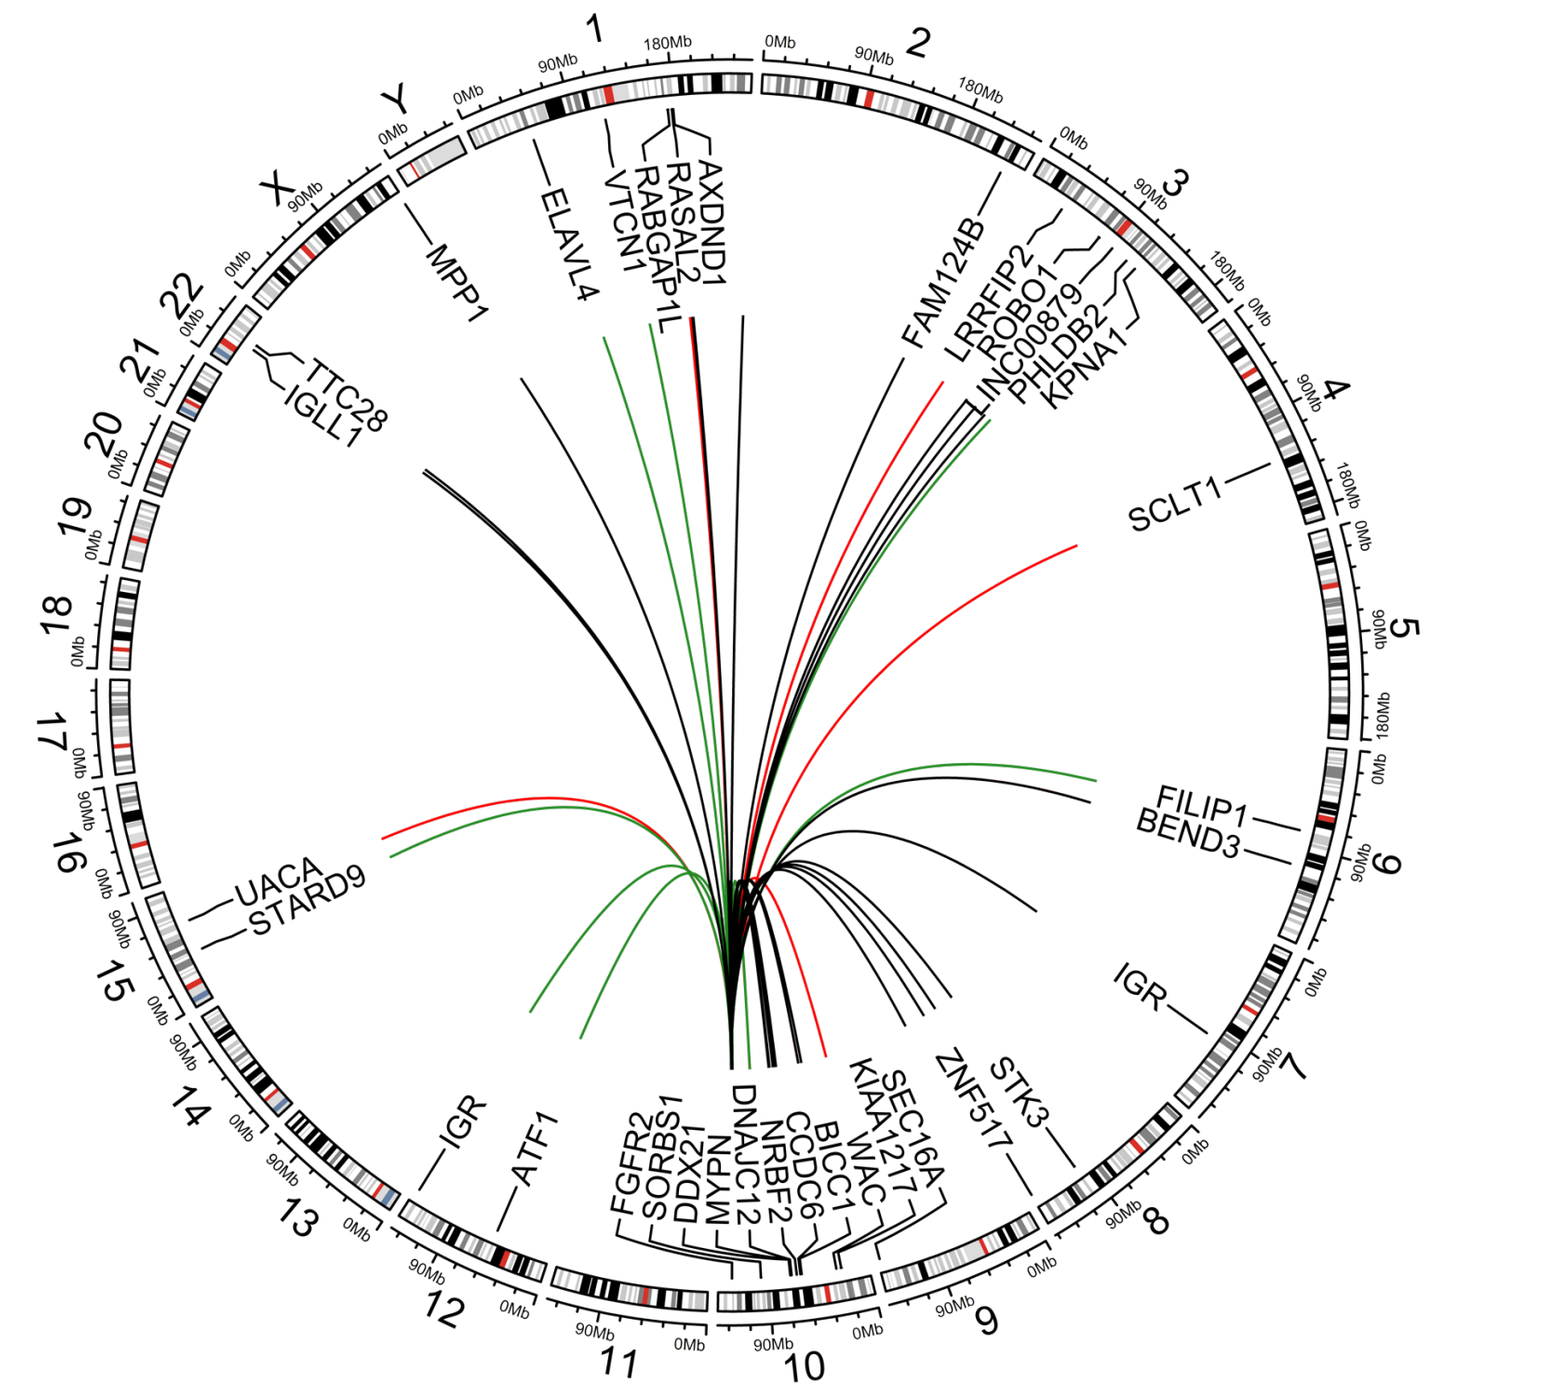
ELAVL4
VTCN1
RABGAP1L
RASAL2
AXDND1
MPP1
TTC28
IGLL1
FAM124B
LRRFIP2
ROBO1
LINC00879
PHLDB2
KPNA1
SCLT1
FILIP1
BEND3
IGR
UACA
STARD9
IGR
ATF1
FGFR2
SORBS1
DDX21
MYPN
DNAJC12
NRBF2
CCDC6
BICC1
WAC
KIAA1217
SEC16A
ZNF517
STK3

Black lines: Translocations identified in both tissue and liquid biopsies

Red lines: Fusions detected in tissue only

Green lines: Translocations detected in liquid biopsies only

N=68

Credit: Circlize r package[7]

Location of *FGFR2* fusions and partner chromosomes

Analysis of tumour tissue and liquid biopsies

- As shown in **Figure 5**, *FGFR2* f/r partners were predominantly located in chromosome 10 (38/68; 55.9%) and chromosome 1 (6/68; 8.8%)
- Nearly half of the *FGFR2* fusion partners identified in this study were unique and/or intergenic DNA (30/68; 44.1%)
- *FGFR2* f/r included 21 *FGFR2-BICC1* fusions and 47 non-*FGFR2-BICC1* fusions

Figure 6. *FGFR2* fusion partners (liquid biopsies)

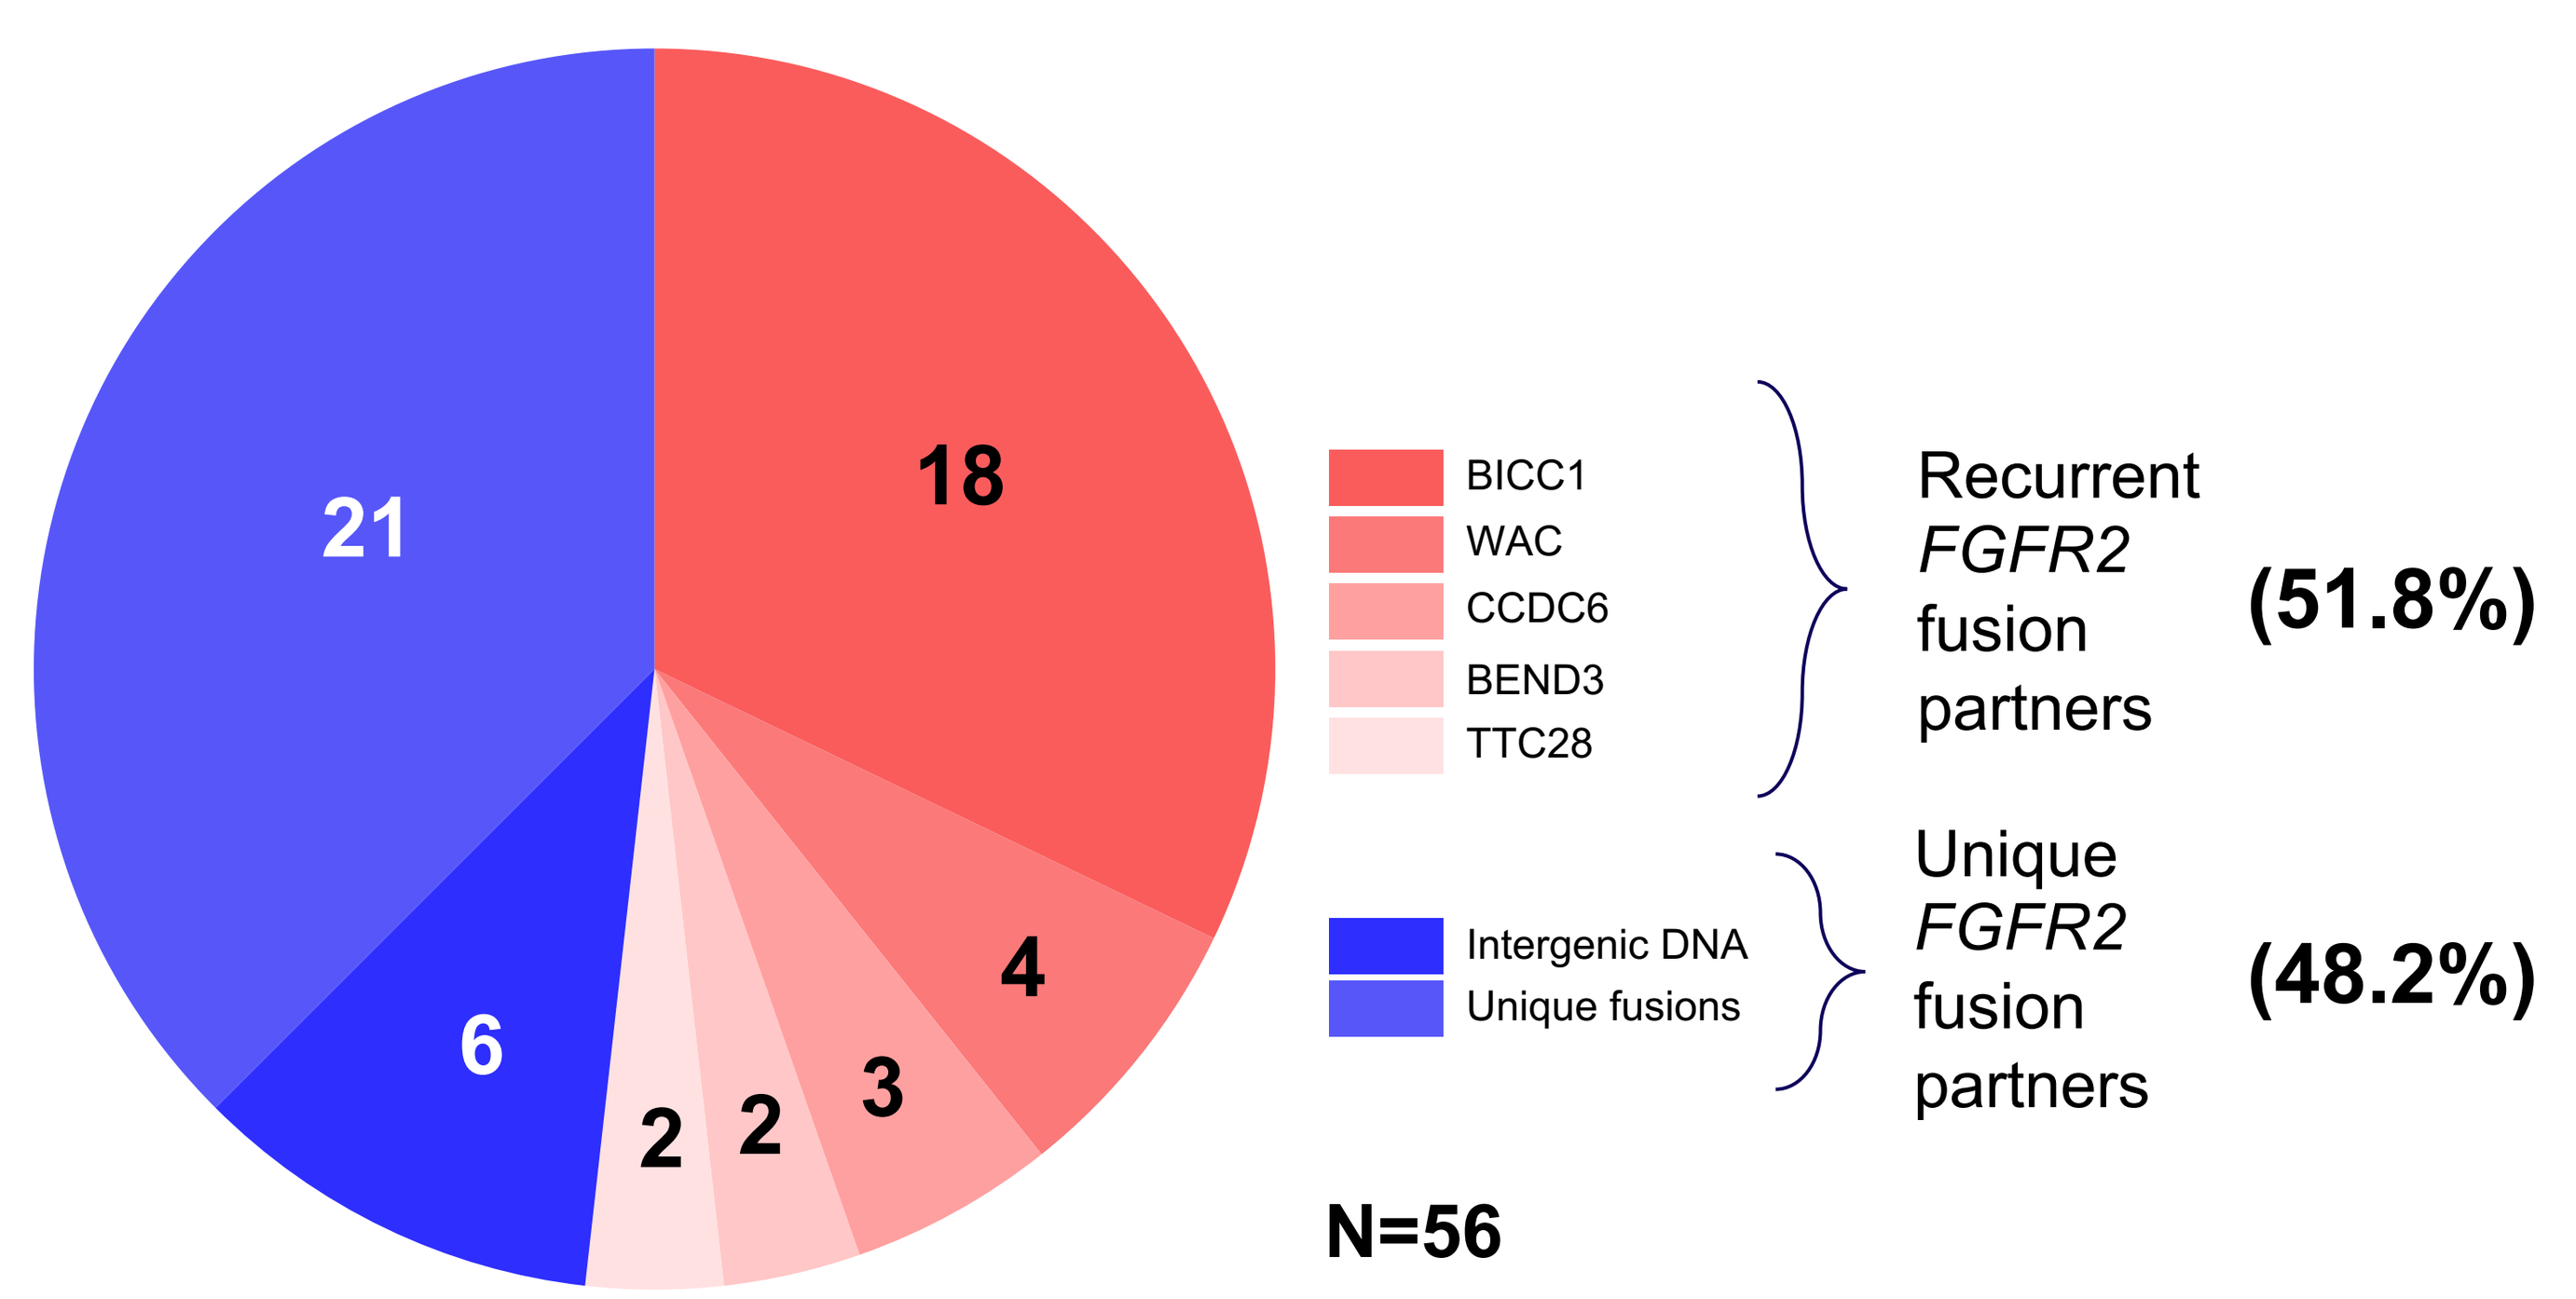
18
4
3
2
2
6
21
BICC1
WAC
CCDC6
BEND3
TTC28
Recurrent *FGFR2* fusion partners
(51.8%)
Intergenic DNA
Unique fusions
Unique *FGFR2* fusion partners
(48.2%)
N=56

Analysis of liquid biopsies

- As shown in **Figure 6**, the predominant *FGFR2* fusion partner gene was *BICC1* (18/56; 31.2%)
- The next most common fusion event seen in liquid biopsies was fusion with intergenic DNA (6/56; 10.7%)
- About half of the *FGFR2* fusion partners were unique (27/56; 48.2%)

CONCLUSIONS

- The fusion partner agnostic *FGFR2* f/r calling algorithm is superior to the standard algorithm in cfDNA, identifying approximately a third of fusions that would otherwise have been missed
- The majority of *FGFR2* f/r detected by the partner agnostic algorithm were unique
- Liquid biopsy NGS identified a high proportion of *FGFR2* f/r, with a rate comparable to tissue NGS
- cfDNA testing offers a rapid and non-invasive mechanism for the detection of resistance mutations
- These encouraging results suggest a role for liquid biopsy in *FGFR2* f/r profiling, providing a non-invasive option to identify *FGFR2* f/r-positive CCA patients
- The role of cfDNA in prospective *FGFR2* f/r profiling will be validated in the ReFocus clinical study, which includes patients with CCA and other solid tumours

References

1. Goyal L, et al. *Cancer Treat Rev.* 2021;95:102170.
2. Yablonovitch A, et al. *J Clin Oncol.* 2020;38(Suppl. 15): Abstract 3545 and poster.
3. Schram AM, et al. *J Clin Oncol.* 2021;39(Suppl. 15): Abstract TPS4165 and poster.
4. Casaletto J, et al. *Cancer Res.* 2021;81(Suppl. 13): Abstract 1455 and poster.
5. Hollebecque A, et al. Oral presentation at ESMO 2022: LBA12.
6. Gu Z, et. al. *Bioinformatics.* 2016;32:2847–9.
7. Gu Z, et. al. *Bioinformatics.* 2014;30:2811–2.

Acknowledgments

The authors would like to thank the patients and their families, all study investigators, sub-investigators, and research staff at participating institutions. The authors also gratefully acknowledge Kai Yu Jen, PhD, Relay Therapeutics, for her contribution to this research, and Guardant Health for use of Guardant360® to enable this work.
Medical writing support was provided by Christine Elsner of BOLDSCIENCE Inc. funded by Relay Therapeutics.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the ENA Symposium and the author of this poster.

Presented at the 34th EORTC-NCI-AACR Symposium, 26–28 October 2022, Barcelona, Spain. For more information, please contact: ClinicalTrials@relaytx.com.