A food effect and esomeprazole drug–drug interaction study of RLY-4008, a highly selective FGFR2 inhibitor, in healthy subjects

Jinshan Shen[1], Rishikesh Sawant[1], Rick Blakesley[1], Kai Yu Jen[1], Fabien Ricard[1], Xiaoyan Li[1], Cassandra Key[2], Djuro Karanovic[1]

[1]Relay Therapeutics, Cambridge, Massachusetts, USA; [2]ICON plc, San Antonio, Texas, USA

Poster No. 2795
Presented at the American Association for Cancer Research (AACR)
Annual Meeting, April 14–19, 2023, Orlando, FL, USA

INTRODUCTION

- The approval of pan-FGFR inhibitors (pan-FGFRi) in cholangiocarcinoma (CCA)[1–3] provides clinical proof-of-concept of FGFR2 as a therapeutic target
- However, the long-term benefit of pan-FGFRi has been limited owing to the emergence of *FGFR2* resistance mutations and side effects that may prevent optimal dosing[4–10]
- RLY-4008, a highly selective, irreversible, orally available small-molecule FGFR2i with broad mutational coverage, was developed by leveraging differences in the conformational dynamics between FGFR2 and other FGFRs
- RLY-4008 has shown promising preliminary efficacy in FGFRi-naïve patients with CCA bearing *FGFR2* fusions/rearrangements[11]
- This study investigated the effect of food and esomeprazole on the pharmacokinetics (PK) of RLY-4008 in healthy subjects

METHODS

Study design

- This was a Phase I, randomized, open-label, three-period, fed and fasted crossover, and drug–drug interaction (DDI) study
- Twenty-four healthy subjects (18–55 years of age) were randomized to receive a single oral dose of RLY-4008 (50 mg) under fasted or fed conditions or with esomeprazole (40 mg) under fasted conditions in one of two sequences (**Figure 1**):
 - Fasting–Fed–DDI
 - Fed–Fasting–DDI

Figure 1. Study design

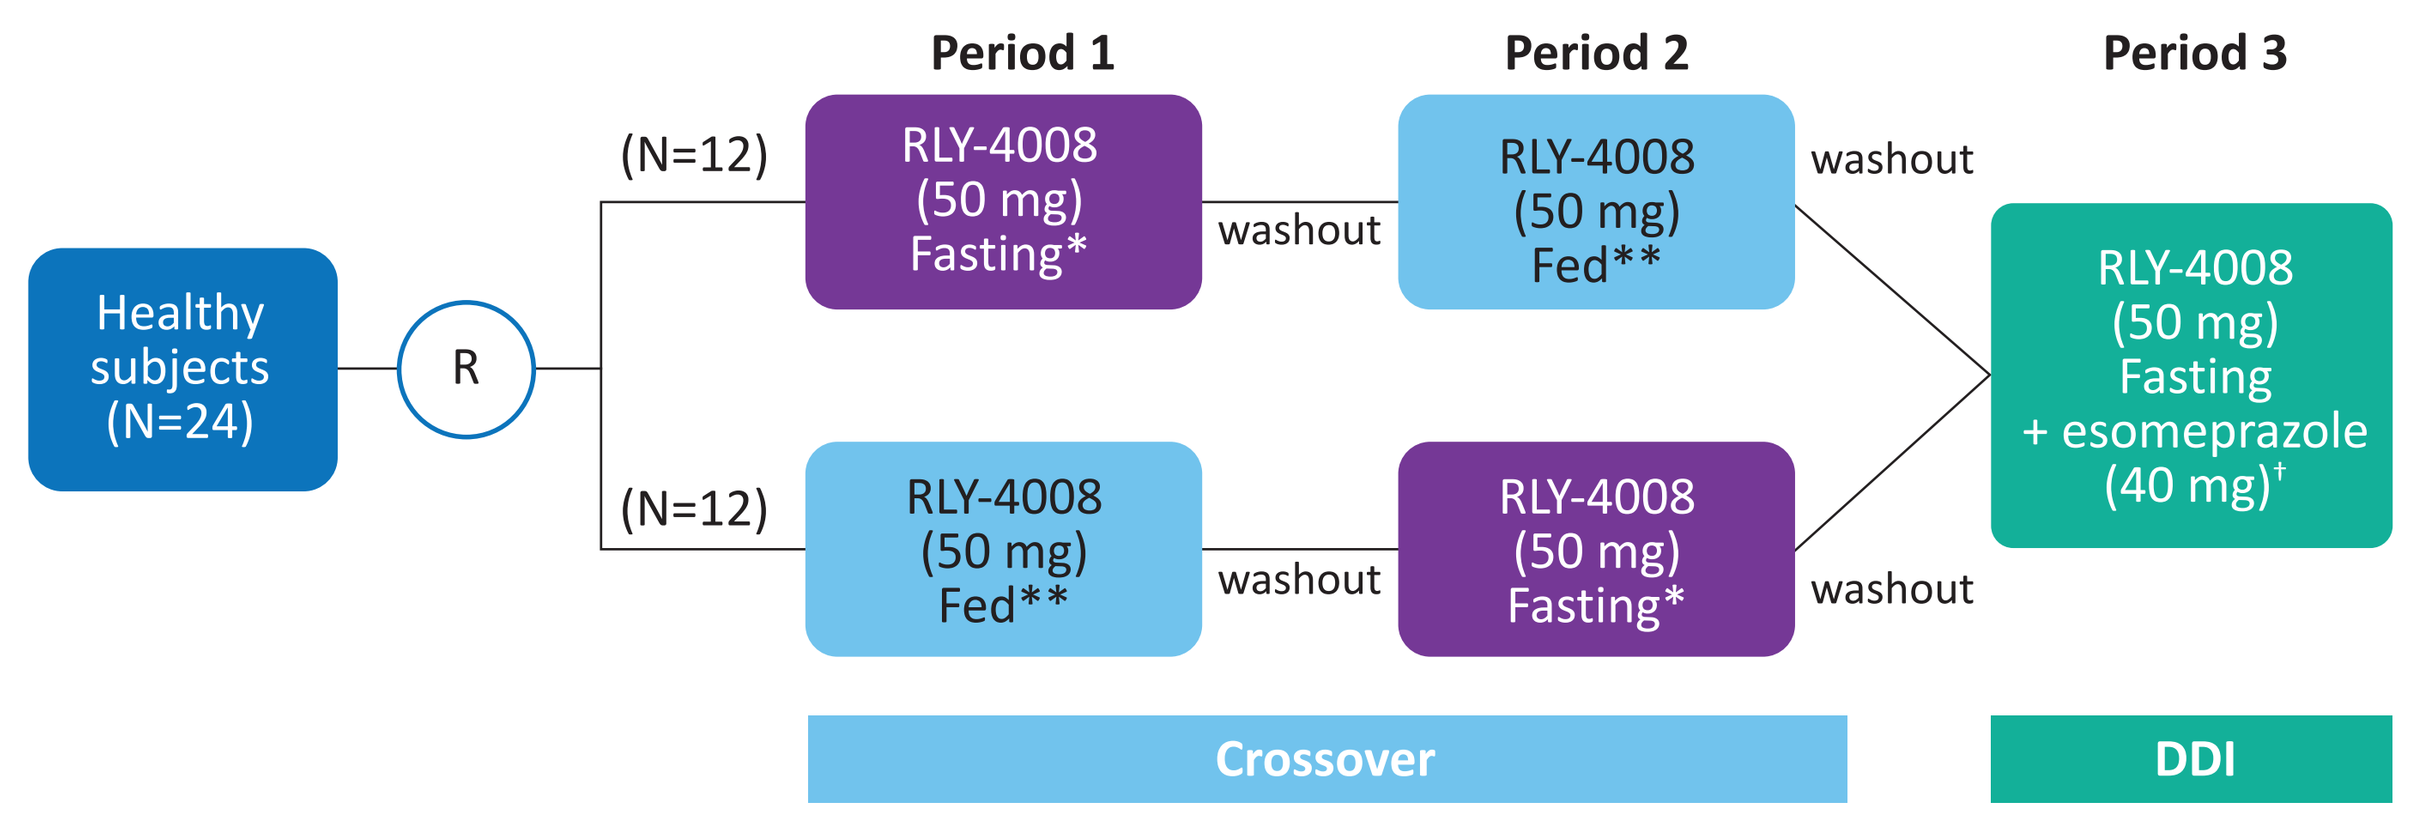
Period 1
Period 2
Period 3
Healthy subjects (N=24)
R
(N=12)
(N=12)
RLY-4008 (50 mg) Fasting*
RLY-4008 (50 mg) Fed**
RLY-4008 (50 mg) Fed**
RLY-4008 (50 mg) Fasting*
washout
washout
washout
washout
RLY-4008 (50 mg) Fasting + esomeprazole (40 mg)†
Crossover
DDI

No food was permitted for 4 hours post-dose. Each of the three study periods included a washout period of ≥7 days.
RLY-4008 PK was assessed up to 120 hours post-dose.
*For the fasted condition, RLY-4008 was administered after an overnight fast of ≥10 hours.
**For the fed condition, subjects received RLY-4008 after a high-fat, high-calorie meal (~800–1000 kcal).
†In the DDI part of the study, esomeprazole was administered alone to fasted subjects for 5 days; on Day 6, it was given to fasted subjects approximately 1 hour before the RLY-4008 dose.
DDI, drug–drug interaction; R, randomization.

Objectives

- The primary objectives were to assess the effect of food or esomeprazole on the PK of RLY-4008 in healthy subjects
- The secondary objective was to evaluate the safety and tolerability of RLY-4008 with and without co-administration of esomeprazole

Assessments

- Blood samples for PK assessments were collected at 0 (pre-dose), 0.5, 1, 1.5, 2, 4, 8, 12, 24, 48, 72, 96, and 120 hours post-RLY-4008 dosing
- Plasma concentrations of RLY-4008 were determined using a validated liquid chromatography–mass spectrometry assay
- Safety and tolerability were assessed based on adverse events (AEs; graded according to Common Terminology Criteria for Adverse Events version 5.0), clinical laboratory results, vital signs, and physical examination

Statistical analyses

- Considering an estimated intra-participant coefficient of variability of 25% and a true mean ratio of 1.00 for RLY-4008 PK parameters, 21 subjects were required to ensure a 90% confidence interval (CI) within an 80–125% range for the mean ratio of area under the concentration–time curve (AUC) and maximum concentration (C_{max}) between the test (RLY-4008 fed or with esomeprazole) and reference (RLY-4008 fasted)
 - Twenty-four subjects were enrolled to account for potential drop-out
- PK variables were calculated from the plasma concentrations data using noncompartmental methods (Phoenix™ WinNonlin® Version 8.0.0.3176, Certara, Princeton, New Jersey, USA)
- A linear mixed-effects model was fitted to log-transformed PK parameters, including C_{max} and AUC. Point estimates and associated 90% CI of geometric least squares mean ratios (LSGMR) were calculated to evaluate the difference between test and reference datasets

RESULTS

Study participants

- Overall, 24 healthy subjects were enrolled, randomized, and completed the study
- Subject demographics and baseline characteristics are shown in **Table 1**

Table 1. Subject demographics and baseline characteristics

	Fasting–Fed–DDI (N=12)	Fed–Fasting–DDI (N=12)	Total (N=24)
Age, median (range), years	38.0 (27–51)	32.5 (22–55)	36.5 (22–55)
Male, n (%)	8 (66.7)	7 (58.3)	15 (62.5)
Ethnicity, n (%)			
Hispanic/Latino	8 (66.7)	6 (50.0)	14 (58.3)
Not Hispanic/Latino	4 (33.3)	6 (50.0)	10 (41.7)
Race, n (%)			
Black or African American	3 (25.0)	6 (50.0)	9 (37.5)
White	8 (66.7)	5 (41.7)	13 (54.2)
Other	1 (8.3)	1 (8.3)	2 (8.3)
Weight, median (range), kg	76.4 (54.0–85.0)	73.1 (56.7–91.1)	75.2 (54.0–91.1)
BMI, mean (SD), kg/m²	25.1 (2.5)	25.5 (2.9)	25.3 (2.7)

BMI, body mass index; DDI, drug–drug interaction; SD, standard deviation.

Effect of food on the PK of RLY-4008

- The median time to maximum concentration (T_{max}) of RLY-4008 was similar when administered under fasted (4.0 h) or fed (4.0 h) conditions
- LSGMRs (90% CI) between RLY-4008 administered with food and under fasting conditions were 87% (77–97%) for C_{max}, 105% (100–110%) for AUC from time 0 to time of the last quantifiable concentration ($AUC_{0-tlast}$), and 106% (100–110%) for AUC from time 0 extrapolated to infinity (AUC_{0-inf}) (**Table 2, Figure 2**)
- The LSGMRs (90% CI) of AUCs were fully contained within the 80–125% range
- The lower bound of 90% confidence of C_{max} LSGMR was below 80%, but the 13% reduction in RLY-4008 C_{max} with food was not considered clinically relevant

Table 2. Effect of food on the PK of RLY-4008 (PK evaluable population; N=24)

PK parameter	Food condition	Geometric LS mean	Comparison of fed vs. fasted: Ratio	Comparison of fed vs. fasted: 90% CI
C_{max} (ng/mL)	Fasted	2481	0.87	0.77–0.97
	Fed	2148		
$AUC_{0-tlast}$ (h*ng/mL)	Fasted	42245	1.05	1.00–1.10
	Fed	44408		
AUC_{0-inf} (h*ng/mL)	Fasted	42452	1.06	1.00–1.10
	Fed	44612		

AUC, area under the plasma concentration–time curve; AUC_{0-inf}, AUC from time 0 extrapolated to infinity; $AUC_{0-tlast}$, AUC from time 0 to time of the last quantifiable concentration; CI, confidence interval; C_{max}, maximum observed plasma concentration; LS mean, least squares mean; PK, pharmacokinetic.

Figure 2. Mean (SD) plasma concentrations of RLY-4008 following a single dose of 50 mg in healthy subjects fed vs. fasted

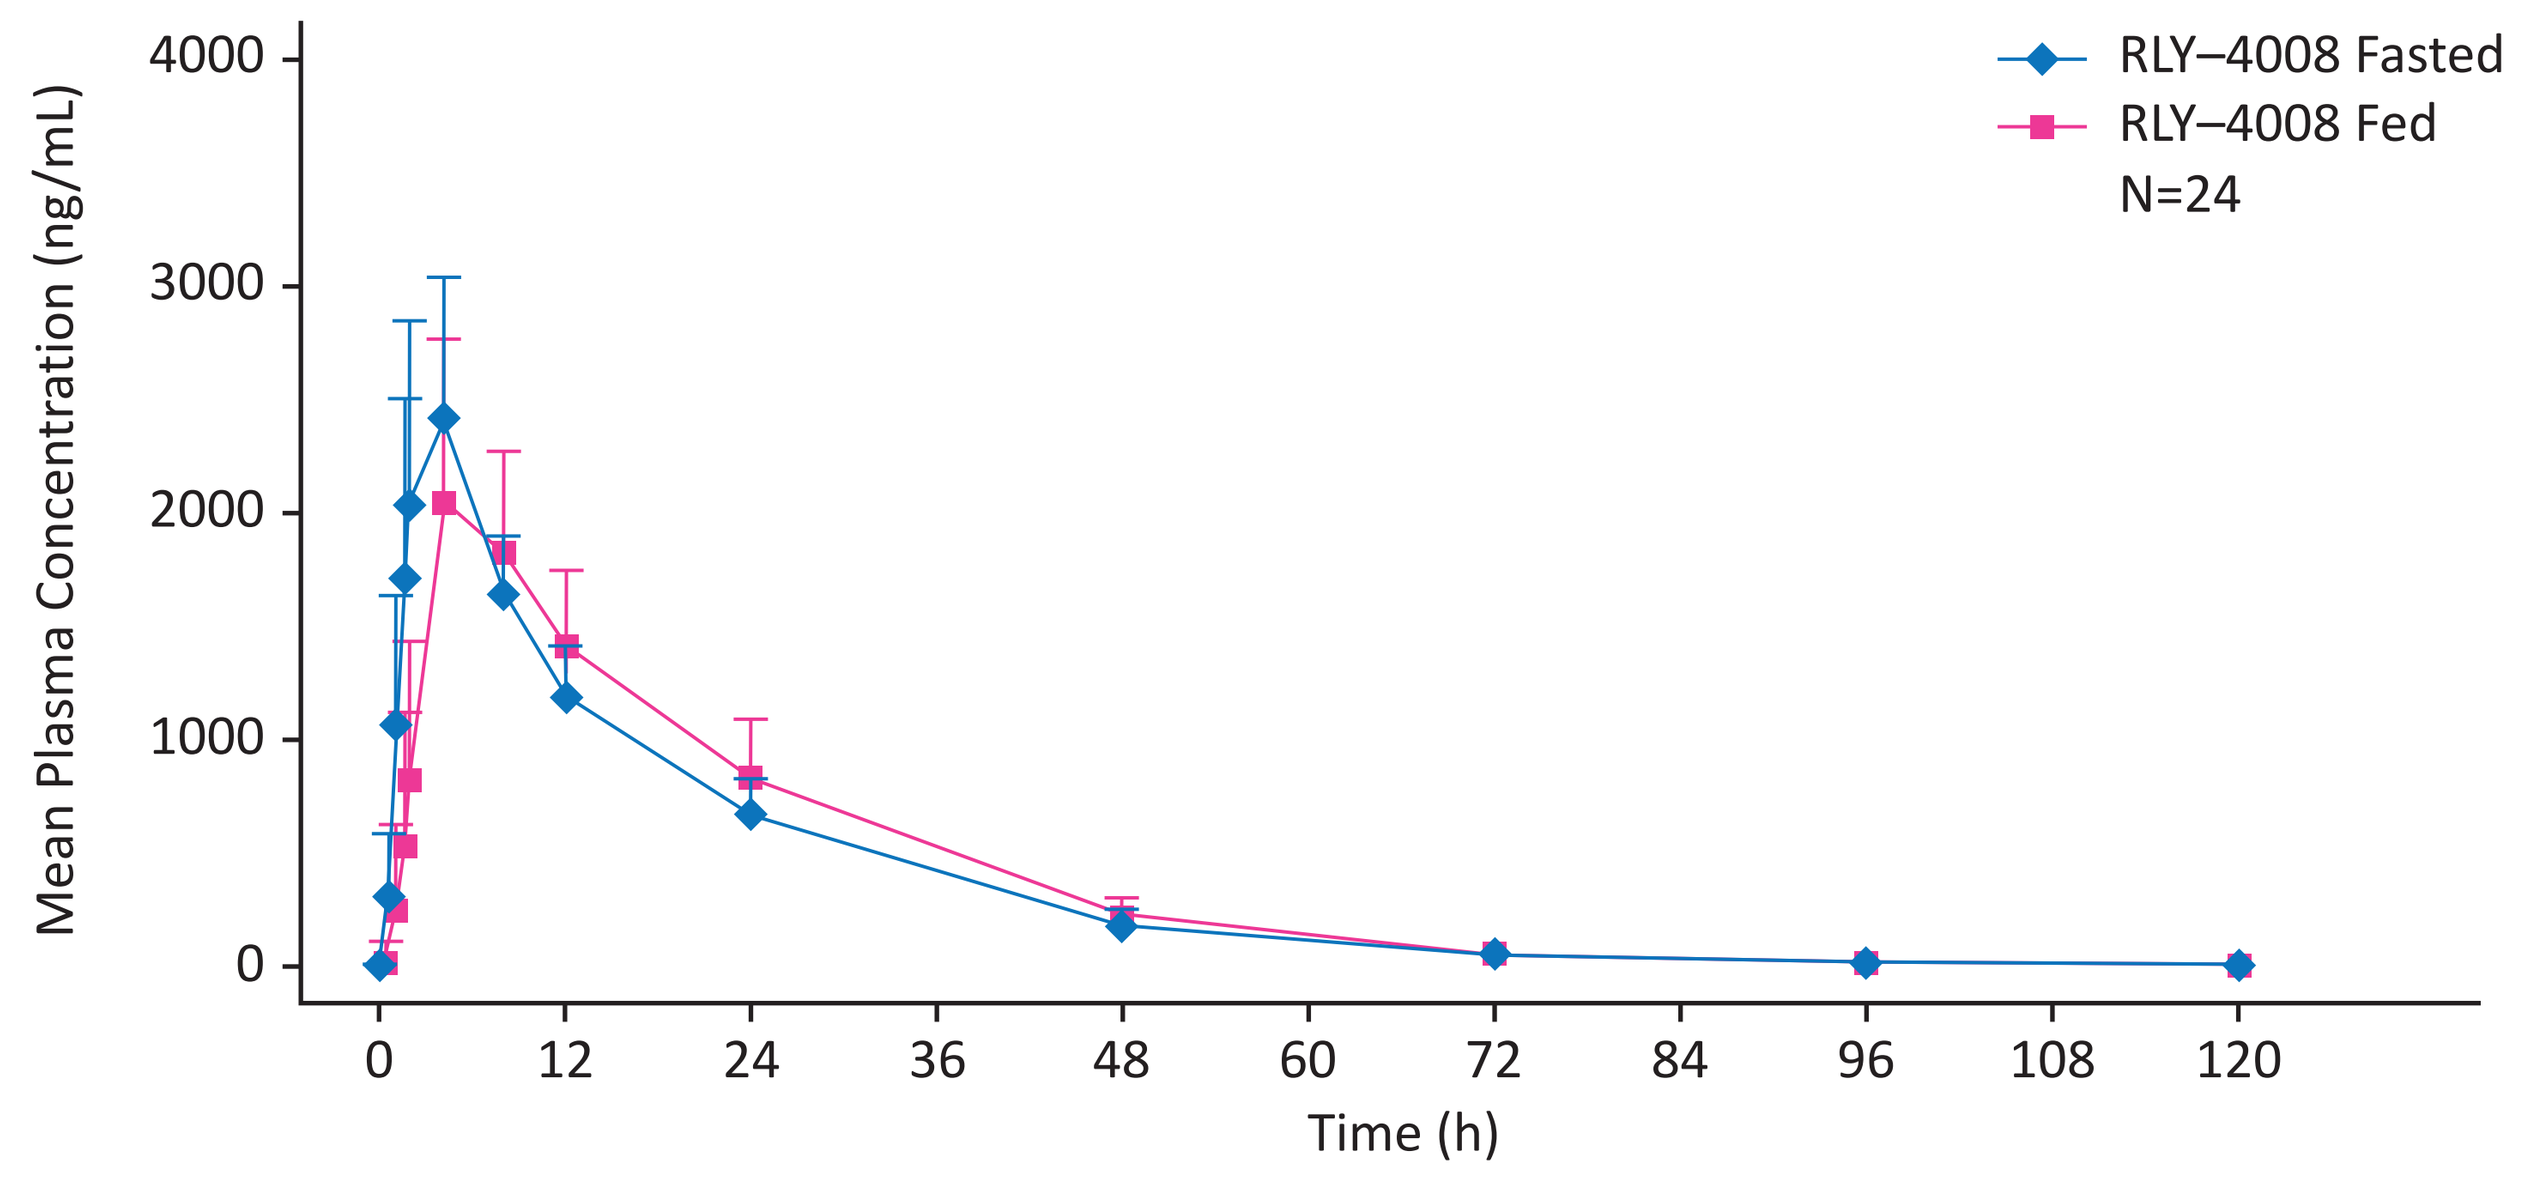
Mean Plasma Concentration (ng/mL)
4000
3000
2000
1000
0
0
12
24
36
48
60
72
84
96
108
120
Time (h)
RLY–4008 Fasted
RLY–4008 Fed
N=24

Points and error bars show the mean + SD of plasma concentrations.
h, hour; SD, standard deviation.

Effect of esomeprazole on the PK of RLY-4008

- The median T_{max} of RLY-4008 was similar when administered under fasted (4.0 h) conditions or fasted conditions with esomeprazole (4.0 h)
- LSGMRs (90% CI) between RLY-4008 administered with esomeprazole and alone, under fasting conditions, were 110% (102–119%) for C_{max}, 113% (109–117%) for $AUC_{0-tlast}$, and 113% (109–117%) for AUC_{0-inf} (**Table 3, Figure 3**)
 - For all parameters, the 90% CIs were fully contained within the 80–125% range

Table 3. Effect of esomeprazole on the PK of RLY-4008 (PK evaluable population; N=24)

PK parameter	Condition	Geometric LS mean	Comparison of fasted + esomeprazole vs. fasted: Ratio	Comparison of fasted + esomeprazole vs. fasted: 90% CI
C_{max} (ng/mL)	Fasted	2481	1.10	1.02–1.19
	Fasted + esomeprazole	2741		
$AUC_{0-tlast}$ (h*ng/mL)	Fasted	42245	1.13	1.09–1.17
	Fasted + esomeprazole	47821		
AUC_{0-inf} (h*ng/mL)	Fasted	42452	1.13	1.09–1.17
	Fasted + esomeprazole	48028		

AUC, area under the plasma concentration–time curve; AUC_{0-inf}, AUC from time 0 extrapolated to infinity; $AUC_{0-tlast}$, AUC from time 0 to time of the last quantifiable concentration; CI, confidence interval; C_{max}, maximum observed plasma concentration; LS mean, least squares mean; PK, pharmacokinetic.

Figure 3. Mean (SD) plasma concentrations of RLY-4008 following a single dose of 50 mg in healthy subjects fasted vs. fasted with esomeprazole

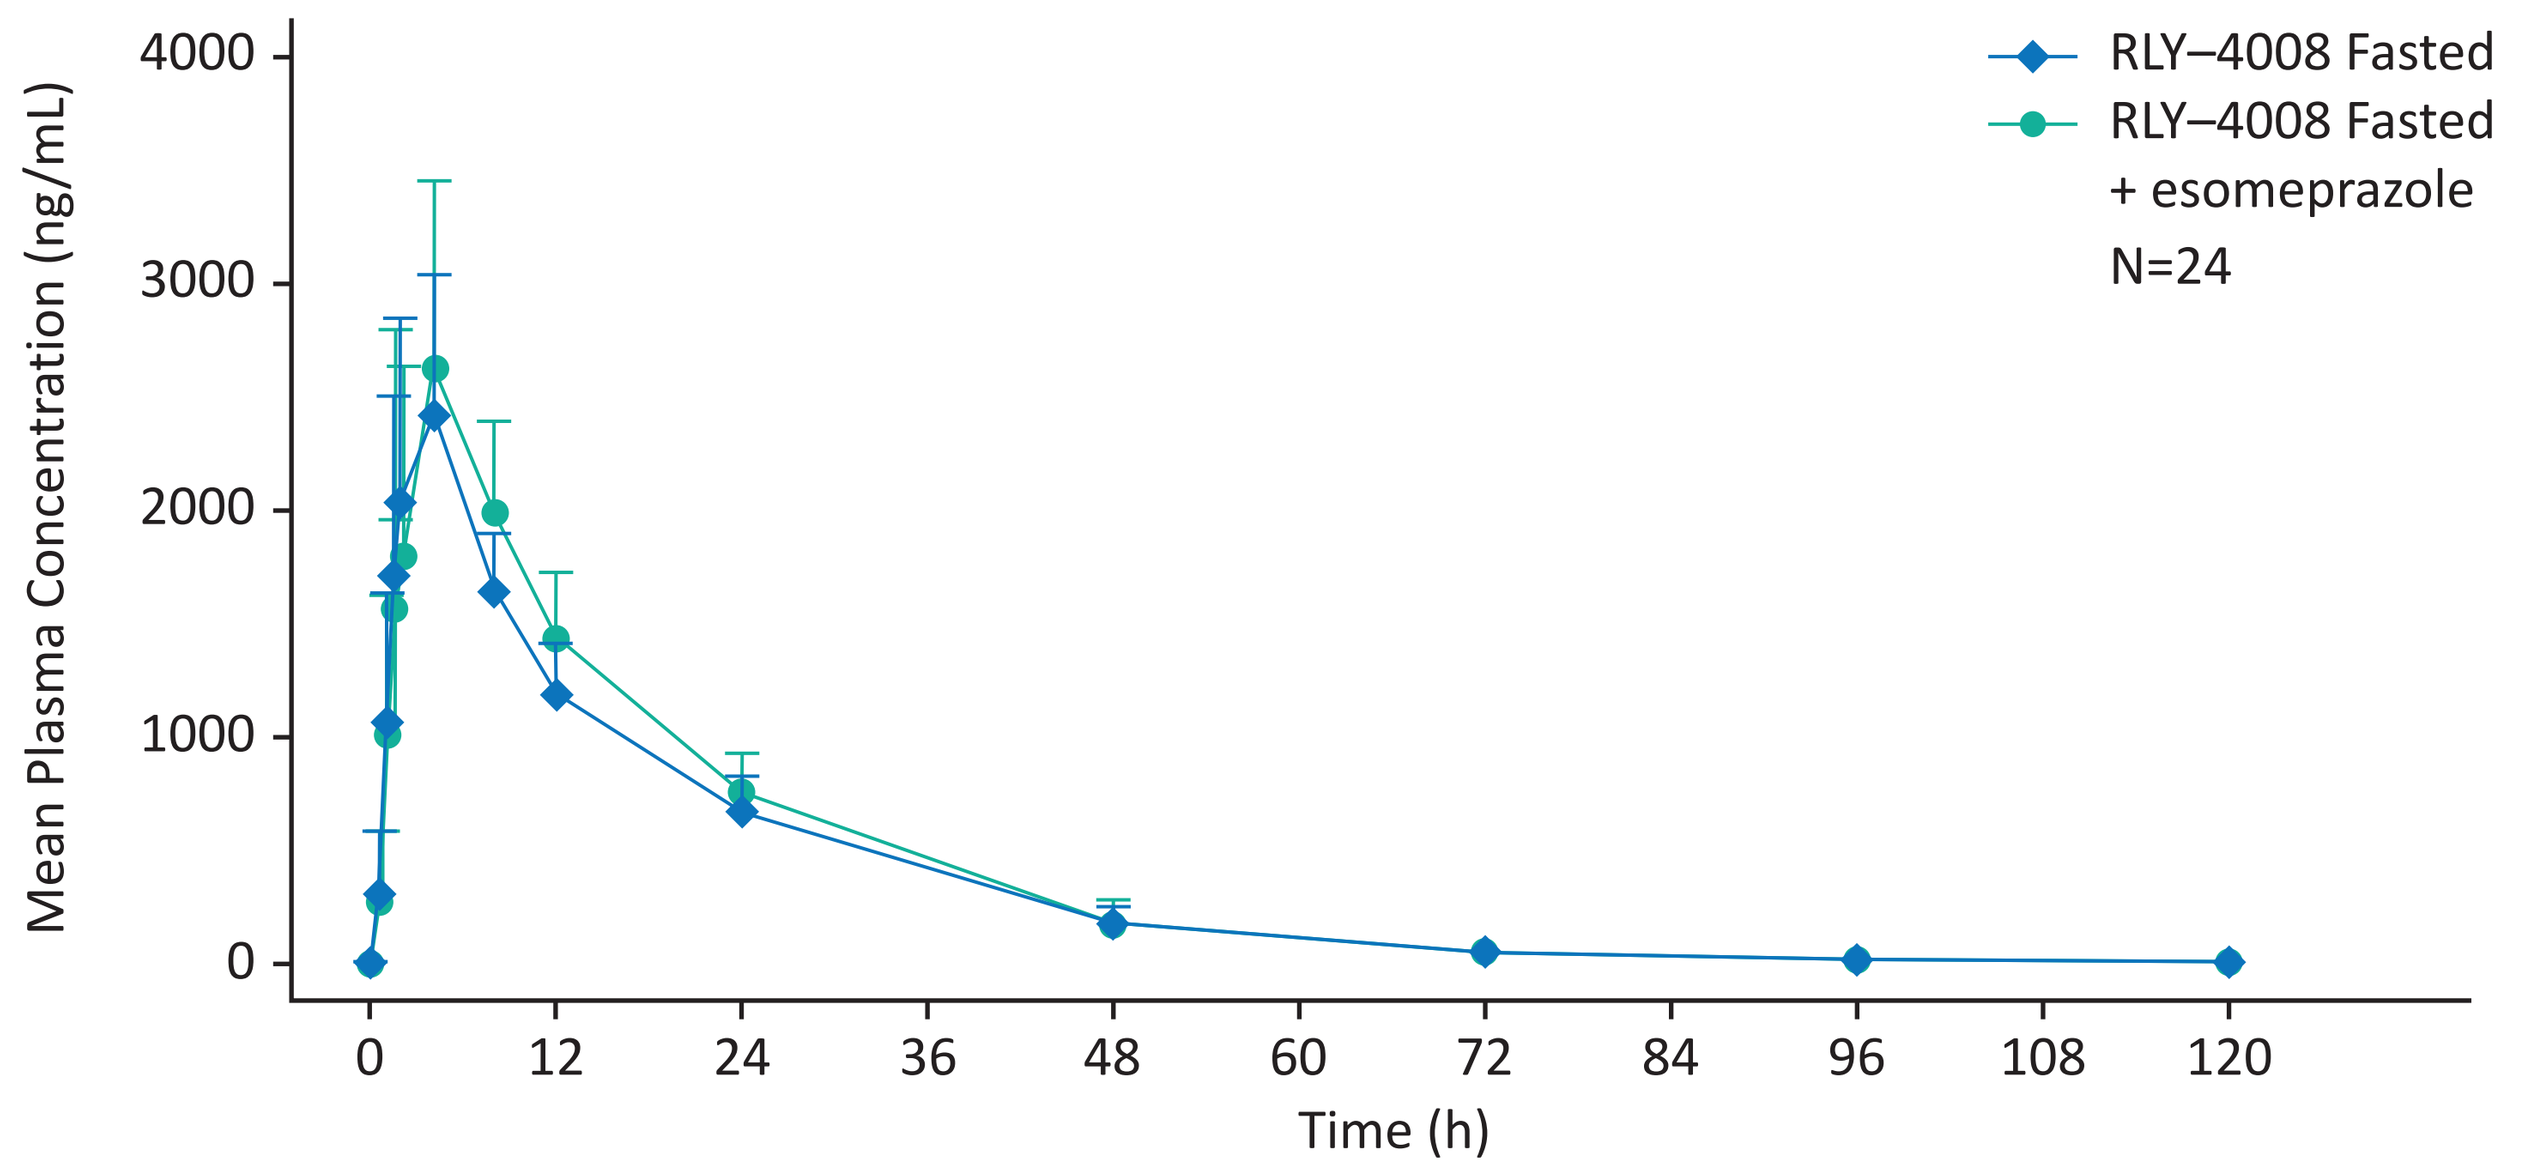
Mean Plasma Concentration (ng/mL)
4000
3000
2000
1000
0
0
12
24
36
48
60
72
84
96
108
120
Time (h)
RLY–4008 Fasted
RLY–4008 Fasted + esomeprazole
N=24

Points and error bars show the mean + SD of plasma concentrations.
h, hour; SD, standard deviation.

Safety and tolerability

- Overall, eight subjects (33.3%) experienced an AE (**Table 4**)
- All AEs were mild/moderate (Grade 1/2) and had an outcome of recovered/resolved. No deaths or serious AEs were reported, and no AEs led to treatment discontinuation

Table 4. Treatment-emergent AEs (safety population)

AE, n (%)	RLY-4008 Fasted (N=24)	RLY-4008 Fed (N=24)	Esomeprazole (N=24)	RLY-4008 (Fasted) + esomeprazole (N=24)	Overall (N=24)
Subjects with any AE	3 (12.5)	2 (8.3)	4 (16.7)	2 (8.3)	8 (33.3)
ALT increase	1 (4.2)	1 (4.2)	0	1 (4.2)	3 (12.5)
AST increase	1 (4.2)	1 (4.2)	0	1 (4.2)	3 (12.5)
Constipation	1 (4.2)	0	1 (4.2)	0	2 (8.3)
Dyspepsia	0	1 (4.2)	1 (4.2)	1 (4.2)	2 (8.3)
Headache	1 (4.2)	0	1 (4.2)	0	2 (8.3)
Skin abrasion	0	1 (4.2)	0	0	1 (4.2)
Arthralgia	0	1 (4.2)	0	0	1 (4.2)
Back pain	0	0	1 (4.2)	0	1 (4.2)

AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase.

CONCLUSIONS

- Food does not have a clinically relevant effect on the PK of RLY-4008 in healthy subjects, indicating that RLY-4008 can be dosed with or without food
- Esomeprazole does not have a clinically relevant effect on the PK of RLY-4008 in healthy subjects, indicating that RLY-4008 can be dosed with or without proton pump inhibitors, histamine type-2 receptor blockers, and antacids
- A single dose of RLY-4008 was well tolerated in healthy subjects when administered fasted, fed, or with esomeprazole
- ReFocus (NCT04526106), a global, open-label Phase 1/2 study of RLY-4008 in adult patients with advanced, FGFR2-driven CCA or other solid tumors, is ongoing

References

1. TRUSELTIQ® (infigratinib). Highlights of prescribing information. Accessed January 2023.
2. PEMAZYRE®(pemigatinib). Highlights of prescribing information. Accessed January 2023.
3. LYTGOBI® (futibatinib). Highlights of prescribing information. Accessed January 2023.
4. Javle M, et al. *Lancet Gastroenterol Hepatol*. 2021;6:803–815.
5. Javle M, et al. *J Clin Oncol*. 2018;36:276–282.
6. Abou-Alfa GK, et al. *Lancet Oncol*. 2020;21:671–684.
7. Goyal L, et al. *J Clin Oncol*. 2022;40(16 Suppl.):4009.
8. Silverman IM, et al. *Cancer Discov*. 2021;11:326–339.
9. Goyal L, et al. *Cancer Discov*. 2017;7:252–263.
10. Goyal L, et al. Presented at the 32nd EORTC/AACR/NCI Virtual Symposium, 2020. Abstract 49.
11. Hollebecque A, et al. *Ann Oncol*; 2022; 33(Suppl. 7):S808–S869.

Acknowledgments

The authors would like to thank the study participants and study investigators. This study was sponsored by Relay Therapeutics, Inc. Medical writing support was provided by Christine Elsner of BOLDSCIENCE Inc., funded by Relay Therapeutics.

Copies of this poster can be obtained through Quick Response Code. These are for personal use only and may not be reproduced without permission from the author of this poster and the AACR.