Mass Balance and Metabolic Profile of Zovegalisib in Male Healthy Subjects

Jinshan Shen,[1] Rishikesh Sawant,[1] Gabe Wolfson,[1] Alison Timm,[1*] Eunice L. Kwak,[1] Kyle Robinson,[2] Stephen English,[3] Taha El-Shahat,[4] Brenton G. Mar[1]

[1]Relay Therapeutics, Cambridge, Massachusetts, USA; [2]Labcorp, Madison, Wisconsin, USA; [3]Pharmaron, Germantown, Maryland, USA; [4]Fortrea, Madison, Wisconsin, USA
*Research conducted while at this institution

Poster number: 704

INTRODUCTION

- Zovegalisib (RLY-2608) is an allosteric, pan-mutant-selective inhibitor that targets mutant forms of phosphatidylinositol 3-kinase alpha (PI3Kα) while sparing wild type
- Relay Therapeutics is developing zovegalisib for the treatment of HR+/HER2− breast cancer and vascular anomalies with *PIK3CA* mutation
- This study was conducted to assess the mass balance, metabolic profiles, pharmacokinetics (PK), and safety of zovegalisib in healthy participants

METHODS

- **Study design and objectives**
 - This was a Phase 1, open-label, nonrandomized, single-dose, single-center study conducted in healthy male participants
 - The primary objective was to determine the mass balance of zovegalisib
 - The secondary objectives were to determine the metabolic profiles and chemical structures of metabolites and PK of zovegalisib, and to assess the safety and tolerability of a single oral dose of zovegalisib when administered to healthy male adult participants
- **Eligibility and treatment**
 - Eligible participants were healthy males ≥18 and ≤55 years old, with a body mass index ≥18.0 and ≤32.0 kg/m^2, and body weight ≥50 and ≤110 kg
 - All participants received a single oral dose of 600 mg/100 µCi [^{14}C]-zovegalisib after an overnight fasting of at least 10 hours (**Figure 1**)

Figure 1. Study Schema

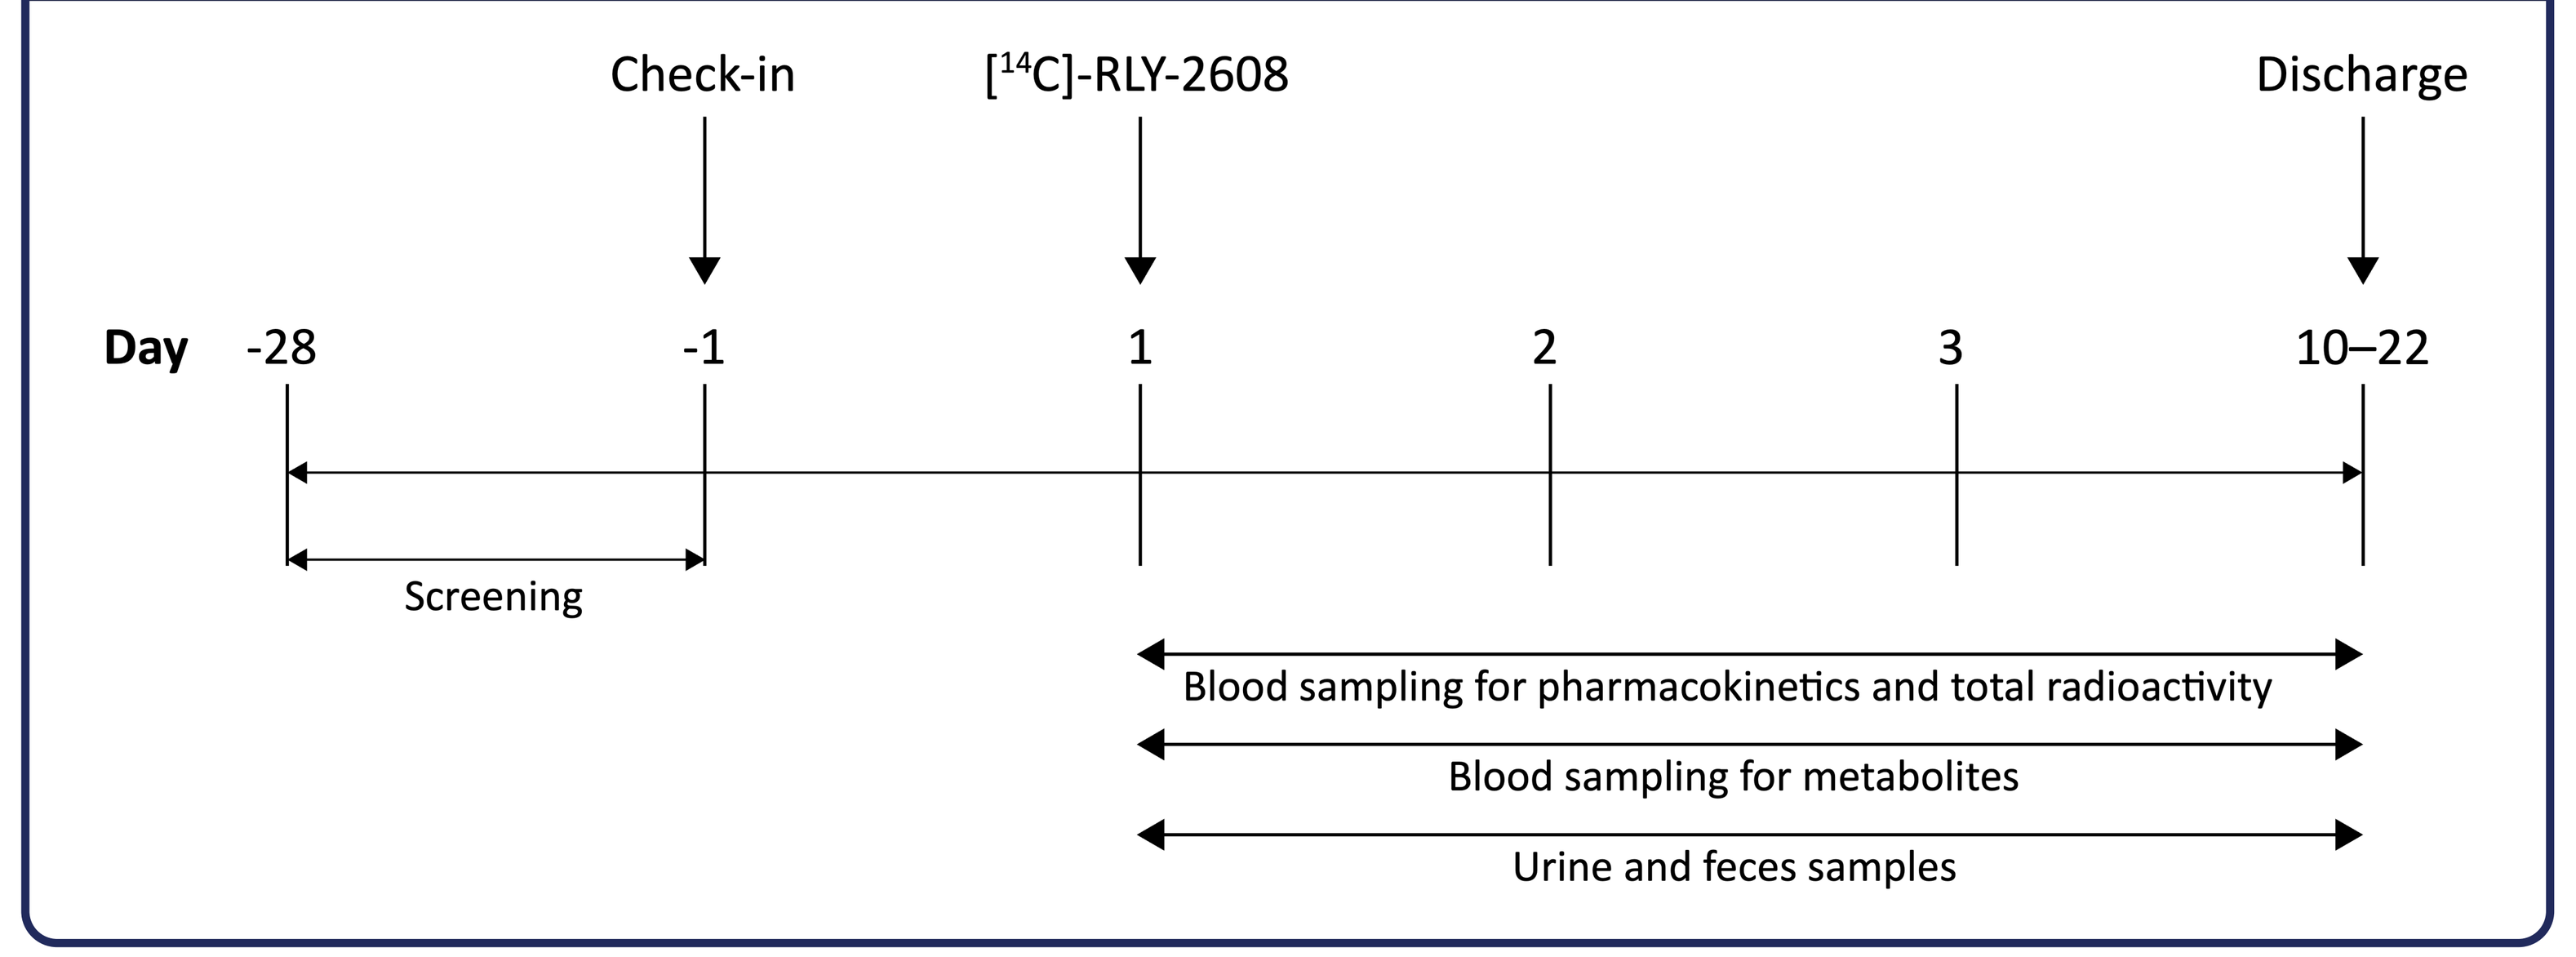
Check-in
[14C]-RLY-2608
Discharge
Day
-28
-1
1
2
3
10–22
Screening
Blood sampling for pharmacokinetics and total radioactivity
Blood sampling for metabolites
Urine and feces samples

- **Assessment**
 - Blood, urine, and feces samples were collected up to 504 hours postdose to assess excretion routes, mass balance, metabolic profiles, and PK of [^{14}C]-zovegalisib
 - [^{14}C]-zovegalisib-related total radioactivity (TRA) was assessed by liquid scintillation counting
 - Metabolic profiling and metabolite identification were assessed by liquid chromatography-high resolution mass spectrometry (LC-HRMS) and accelerator mass spectrometry (AMS)
 - Plasma concentrations of zovegalisib were quantified by a validated LC-MS/MS assay
 - PK parameters were estimated using software Phoenix® WinNonlin (Version 8.3.5, Certara, Radnor, PA)
 - Safety and tolerability were assessed based on adverse events (AEs), as graded according to the National Cancer Institute Common Terminology Criteria for Adverse Events version 5.0, clinical laboratory results, vital signs, and physical examination

RESULTS

Study Participants

- 8 healthy male participants were enrolled and completed the study
- Participant demographics and baseline characteristics are summarized in Table 1

Table 1. Participant Demographics and Baseline Characteristics

Demographic	600 mg [^{14}C]-Zovegalisib (N=8)
Age, years	34.5 (7.62) [23-46]
Sex, n (%)	
Male	8 (100%)
Race, n (%)	
White	4 (50.0%)
Black or African American	3 (37.5%)
Asian	1 (12.5%)
Ethnicity, n (%)	
Not Hispanic or Latino	7 (87.5%)
Hispanic or Latino	1 (12.5%)
Height, cm	173.89 (10.478)
Body weight, kg	81.76 (16.096)
Body mass index, kg/m^2	26.83 (2.864)

Note: For continuous data, mean (SD) presented; for categorical data, n (%) presented; for age, mean (SD) [range] presented.

Mass Balance of [^{14}C]-zovegalisib

- [^{14}C]-zovegalisib-related TRA recovery was 92.8 ± 12.7% of the dose administered (**Figure 2**)
 - Levels of radioactivity in urine were below the limit of quantitation in all evaluable participants
 - All [^{14}C]-zovegalisib-related TRA was recovered in feces, indicating fecal excretion as the major elimination pathway

Figure 2. Mean (± SD) Cumulative Percent of Radioactive Dose Recovered in Feces following a Single Oral Dose of 600 mg/100 µCi [^{14}C]-Zovegalisib

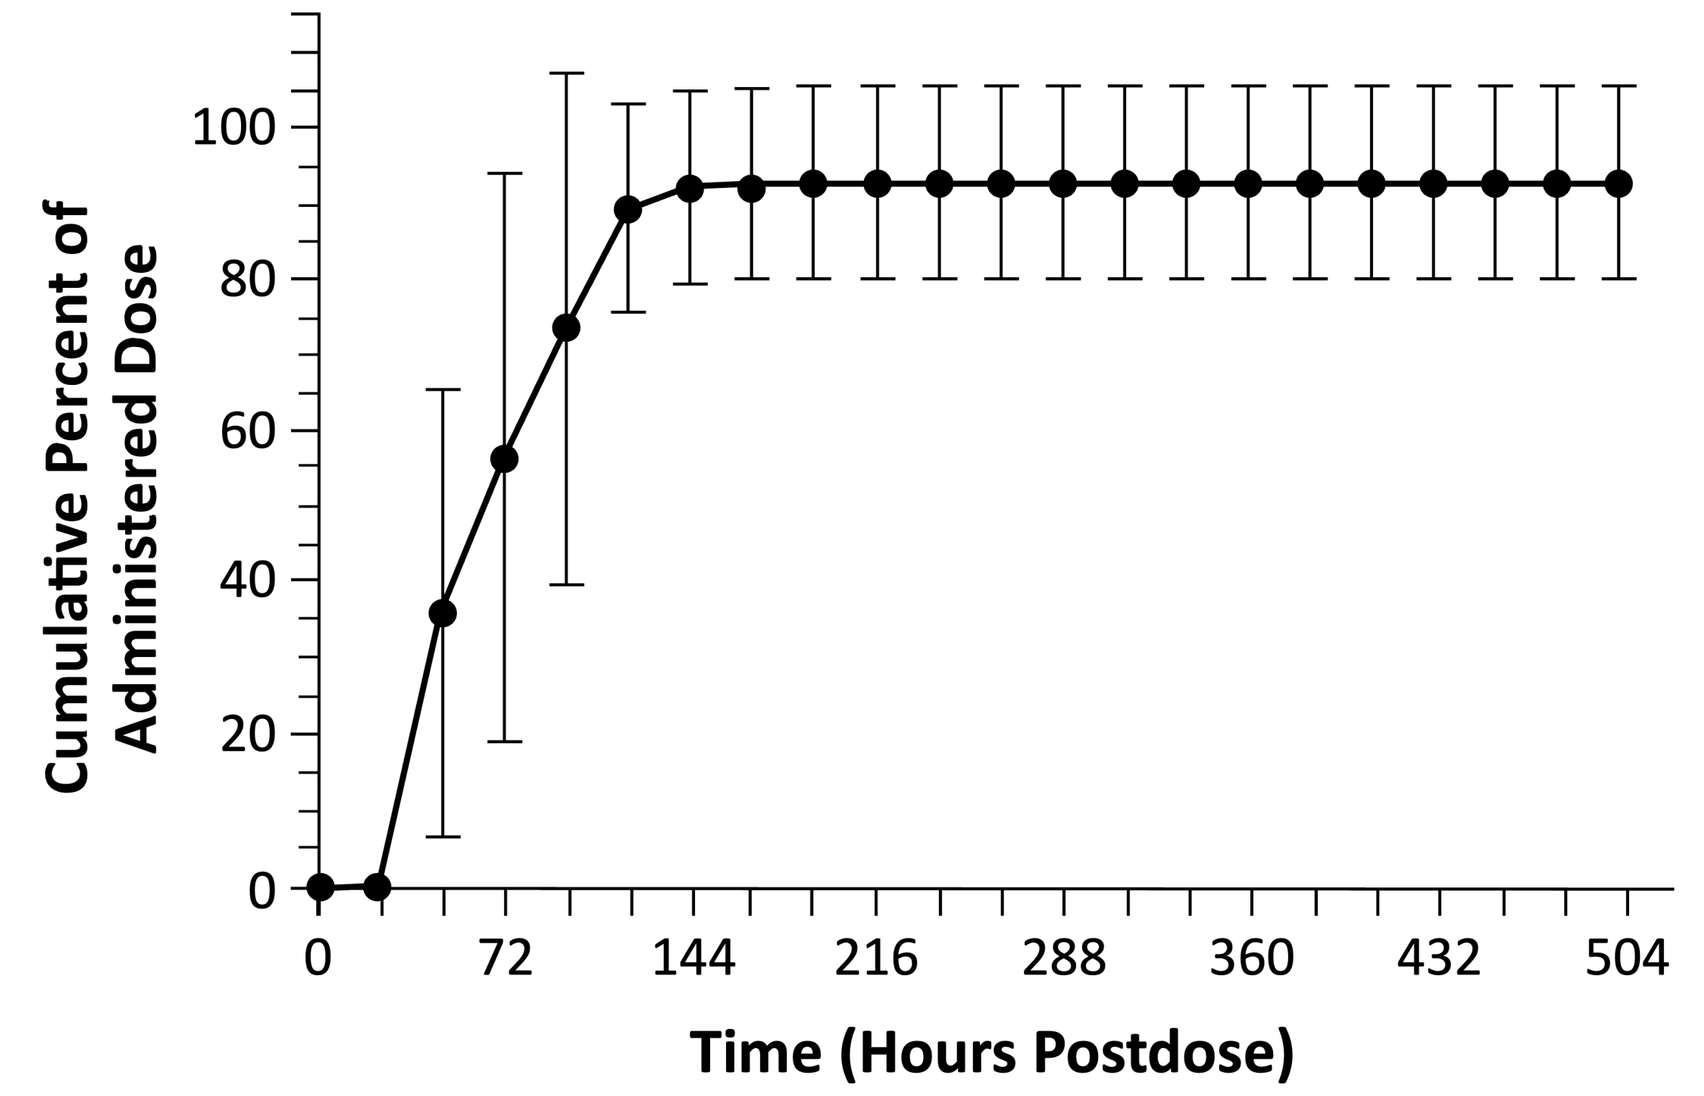
Cumulative Percent of Administered Dose
0
20
40
60
80
100
0
72
144
216
288
360
432
504
Time (Hours Postdose)

Note: 1 study participant was excluded from mass balance and metabolic profiling analyses due to quantifiable radioactivity in the predose urine sample.
SD, standard deviation.

Metabolic Profiling and Metabolite Identification

- Metabolic profiles of [^{14}C]-zovegalisib in pooled plasma samples were generated by LC fractionation followed by AM analysis (**Figure 3A**)
 - Unchanged [^{14}C]-zovegalisib (P2) was identified as the only major radioactive component, accounting for 86.6% of the TRA contained in the $AUC_{(0\text{-}192h)}$ pool,[1] indicating no circulating major metabolite of zovegalisib following a single oral dosing of 600 mg
 - The LC method used for profiling was an achiral method, thus the measured radioactivity of zovegalisib peak represented both zovegalisib and its coeluting enantiomer RTX-4075 (**Figure 4**)
- Metabolic profiles of [^{14}C]-zovegalisib in pooled feces samples were generated by LC fractionation followed by MicroBeta2 analysis (**Figure 3B**)
 - Unchanged [^{14}C]-zovegalisib was identified as the only major radioactive component, accounting for 91.6% of the administered dose, indicating no major metabolite of zovegalisib in excreta following a single oral dose of 600 mg
 - The LC method used for profiling was an achiral method, thus the measured radioactivity of RLY-2608 peak represented both zovegalisib and its coeluting enantiomer RTX-4075
- Structure identification by LC-HRMS indicated that the major radioactive component within the metabolic profiles of plasma and feces sample extractions was zovegalisib and its enantiomer RTX-4075 (**Table 2**)

Figure 3. Radiochromatogram of Pooled Plasma Extract (A) and Pooled Feces (B) After a Single Oral Dose of [^{14}C]-RLY-2608 to Healthy Male Participants

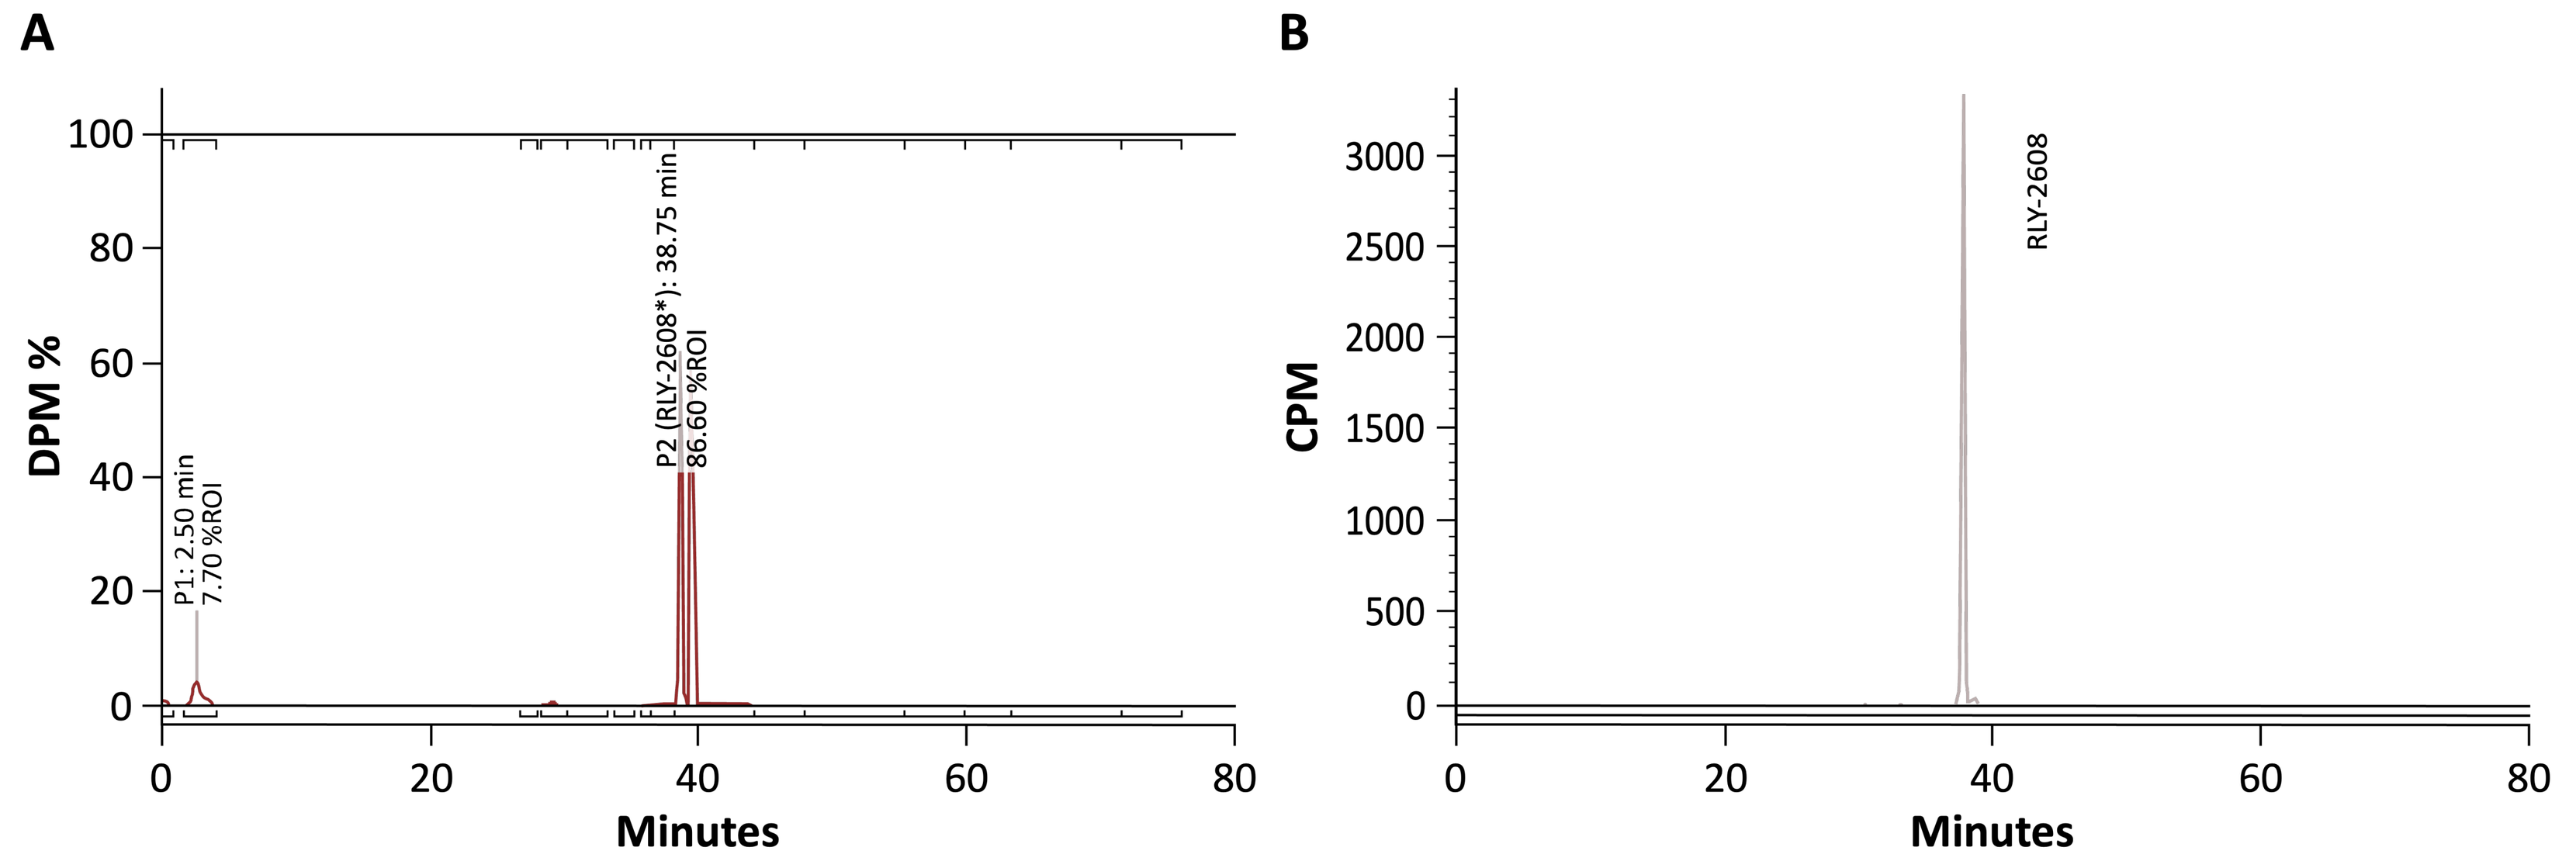
A
DPM %
0
20
40
60
80
100
P1: 2.50 min
7.70 %ROI
P2 (RLY-2608*): 38.75 min
86.60 %ROI
0
20
40
60
80
Minutes
B
CPM
0
500
1000
1500
2000
2500
3000
RLY-2608
0
20
40
60
80
Minutes

Note: Plasma samples were pooled across participants using Hamilton pooling method.[1] Presence of double peaks of P1 was an artefact of the shift of retention time of zovegalisib from 39 minutes to 38 minutes in 2 subsequent injections of 50 µL extract samples, which were both used to generate the radiochromatogram.
CPM, count per minute; DPM, disintegrations per minute; ROI, region of interest.

Note: Representative radiochromatogram from participants 101-204. Fecal samples were pooled within a participant across collection intervals.

Table 2. Identification of Zovegalisib as the Major Radioactive Component Within Plasma and Feces Sample Extracts Through LC-HRMS Analysis

Parent/ Metabolite Designation	Retention Time (Minutes)	[M+H]$^+$ / [M-H]$^-$	Proposed Metabolite Identification	Characteristic Product Ions (m/z)	Matrix
Zovegalisib	37.89[a] 37.85[b]	607 [M-H]$^-$	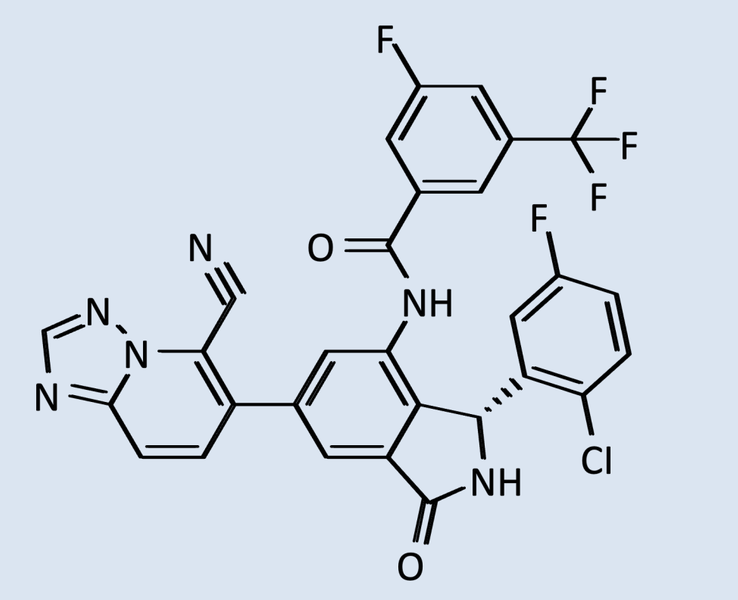	571, 422, 407, 379, 352, 325	Plasma Feces
Zovegalisib	37.87[a] 37.95[b]	609 [M+H]$^+$	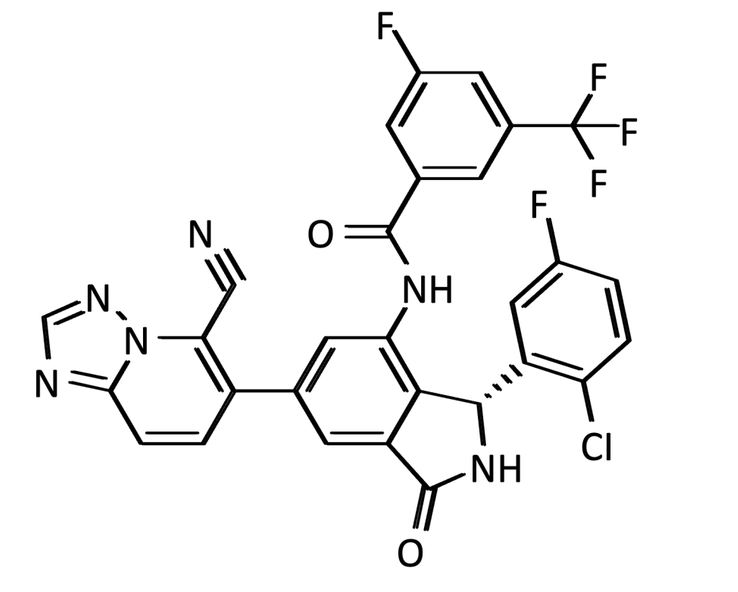	417, 381, 290, 191, 163, 143	Plasma Feces

Note: Because an achiral LC method was used, the zovegalisib signal corresponds to coeluting zovegalisib and its enantiomer RTX-4075.
[a]Retention time from analysis of a representative plasma sample. [b]Retention time from analysis of a representative feces sample.
LC, liquid chromatography; LC-HRMS, LC-high resolution mass spectrometry.

Zovegalisib PK

- Following a single oral dose of 600 mg, zovegalisib was absorbed rapidly, with a median (range) t_{max} of 4.00 h (4.00-5.00 h) (**Figure 4, Table 3**)
- The geometric mean (CV%) C_{max}, $AUC_{0\text{-}\infty}$, and $t_{1/2}$ of zovegalisib were 400 ng/mL (24.4%), 15300 h*ng/mL (39.7%), and 23.6 h (20.2%), respectively
- The geometric mean (CV%) ratio of $AUC_{0\text{-}\infty}$ between enantiomer RTX-4075 and zovegalisib was 0.659 (12.9%)

Figure 4. Mean (+SD) Plasma Concentration Time Profiles of Zovegalisib and Enantiomer RTX-4075

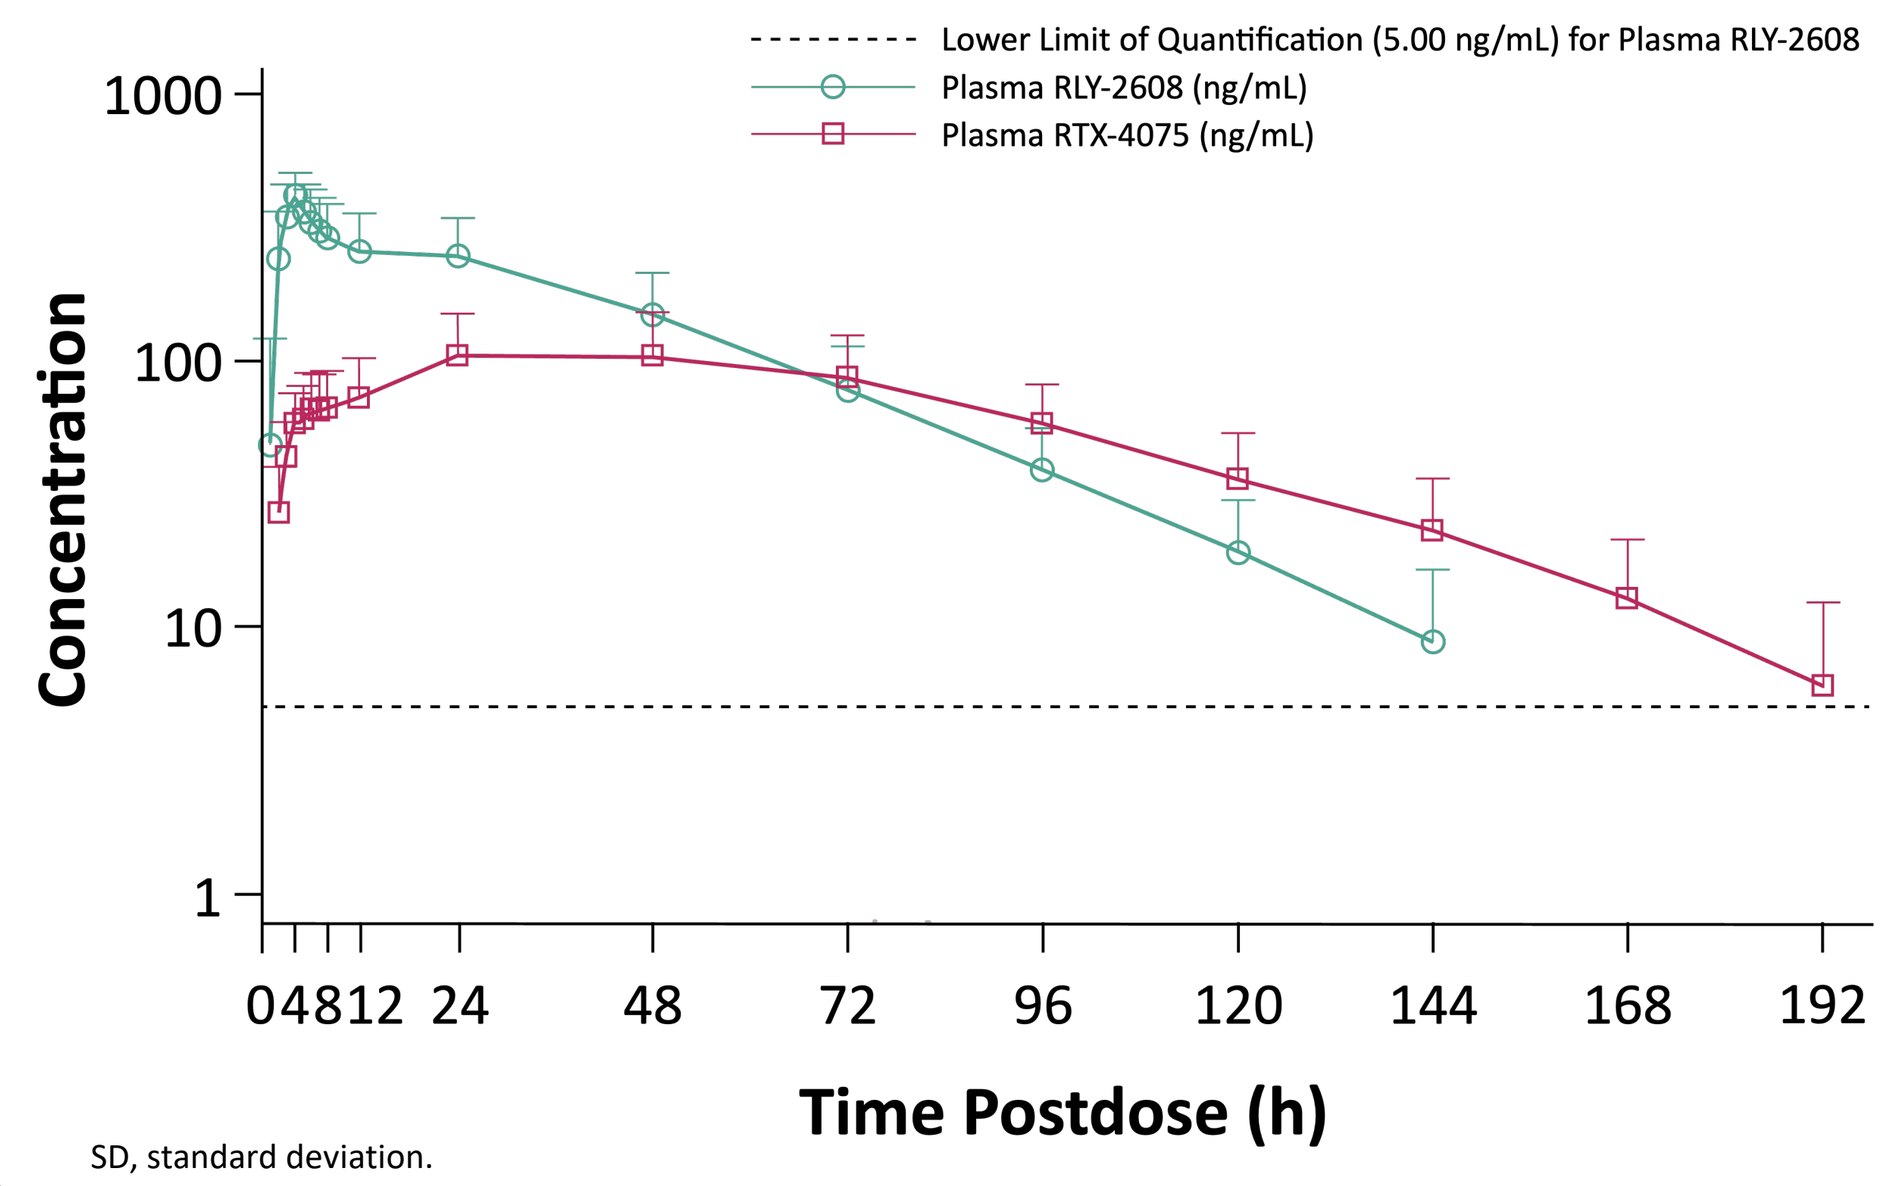
Lower Limit of Quantification (5.00 ng/mL) for Plasma RLY-2608
Plasma RLY-2608 (ng/mL)
Plasma RTX-4075 (ng/mL)
Concentration
1
10
100
1000
04812 24
48
72
96
120
144
168
192
Time Postdose (h)

SD, standard deviation.

Safety and Tolerability

- 4 participants reported treatment-emergent AEs (TEAEs) during the study. All TEAEs were grade 1 and had resolved by the end of the study without intervention
- There were no clinically significant findings in clinical laboratory evaluations, vital signs, or 12-lead electrocardiogram

Table 3. Plasma PK Parameter Estimates of Zovegalisib and Enantiomer RTX-4075 (N=8)

Parameter	Zovegalisib	RTX-4075
$AUC_{0\text{-tlast}}$ (h*ng/mL)	15000 (40.2)	9720 (48.6)
$AUC_{0\text{-}\infty}$ (h*ng/mL)	15300 (39.7)	10100 (47.6)
C_{max} (ng/mL)	400 (24.4)	98.3 (50.2)
t_{max} (h)	4.00 (4.00-5.00)	36.0 (24.0-48.0)
$t_{1/2}$ (h)	23.6 (20.2)	28.8 (21.3)
CL/F (L/h)	40.0 (39.4)	not reported
V_z/F (L)	1360 (49.8)	not reported
$SR_{AUCO\text{-}\infty}$	not reported	0.659 (12.9)

Note: PK parameters are presented as geometric mean (CV%) except T_{max} which is presented as median (range).

CONCLUSIONS

- Following a single oral dose of zovegalisib in humans, fecal excretion was the predominant elimination pathway. No major metabolite of zovegalisib was detected
- Zovegalisib was absorbed rapidly. The $AUC_{(0\text{-}\infty)}$ ratio between enantiomer RTX-4075 and zovegalisib was 0.659
- Single oral dose of 600 mg [^{14}C]-zovegalisib was safe and well tolerated in healthy male participants

Reference

1. Hamilton RA, Garnett WR, Kline BJ. *Clin Pharmacol Ther*. 1981;29:408-413.

Acknowledgments

The authors would like to thank the study participants and their families, and study investigators and research staff. This study is sponsored by Relay Therapeutics, Inc. Medical writing support was provided by Yaeko Hiyama, PhD, of Bio Connections, LLC, funded by Relay Therapeutics.

Abbreviations

AE, adverse event; AMS, accelerator mass spectrometry; AUC, area under the curve; $AUC_{0\text{-}\infty}$, area under the curve from 0 to infinity; $AUC_{0\text{-tlast}}$, AUC from 0 to the last measured time point; CL/F, oral clearance; C_{max}, maximum concentration; CV, coefficient of variation; HER2, human epidermal growth factor receptor 2; HR, hormone receptor; LC, liquid chromatography; LC-HRMS, LC-high resolution mass spectrometry; PIK3CA, phosphatidylinositol-4,5-bisphosphate 3-kinase catalytic subunit alpha; PK, pharmacokinetic; SR, cumulative shear rate; $t_{1/2}$, half-life; T_{max}, time to maximum concentration; TEAE, treatment-emergent AE; TRA, total radioactivity; V_z/F, volume of distribution during terminal phase.

Copies of this poster obtained through QR, AR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at the 2026 American College of Clinical Pharmacy (ACCP) Annual Meeting, 27-29 Sept 2026, North Bethesda, Maryland, USA